P-MUC1C-ALLO1: A Fully Allogeneic Stem Cell Memory T Cell (T_{SCM}) CAR-T Therapy with Broad Potential in Solid Tumor

Yan Zhang, Anna K. Kozlowska, Jacqueline Fritz, Yingying Zhao, Claudia Palomino La Torre, Stacey Cranert, Steven Wang, Rebecca Codde, Elvira Argus, Samad Ibitokou, Vanitra Richardson, Sumiti Jain, Maximilian Richter, Deepak Patil, Yening Tan, Min Tong, Lu Yao, Majid Ghoddusi, Eric M. Ostertag, Julia Coronella, Devon J. Shedlock

Poseida Therapeutics, 9390 Towne Centre Drive, Suite 200, San Diego, CA 92121 USA

ABSTRACT

CAR-T have demonstrated potent activity against hematologic tumors, but less success has been seen against solid tumors. We report generation of T_{SCM}-enriched allogeneic MUC1-C-specific CAR-T cells, P-MUC1C-ALLO1, with potential to treat a broad range of epithelial-derived solid tumors. The proliferative capacity and metabolic profile of T_{SCM} CAR-T are well-suited to activity in the solid tumor setting. MUC1 is comprised of an N-terminal subunit (MUC1-N) tethered to a C-terminal subunit (MUC1-C), forming a stable complex on the cell surface. During tumorigenesis, MUC1 becomes both overexpressed and hypo-glycosylated on many carcinomas. Furthermore, MUC1 undergoes proteolytic cleavage in the tumor microenvironment, leaving behind a proteolytic "stump" of MUC1-C that is over-represented in cancer, making it an attractive therapeutic target.

P-MUC1C-ALLO1 is manufactured using the piggyBac® DNA Delivery System for CAR transgene insertion and the Cas-CLOVER™ Site-Specific Gene Editing System to knockout both the TCR and MHC class I proteins. The addition of a selectable marker within the transposon allows for selection of a fully CAR-positive population, while any residual TCR-positive cells are removed at the end of production by purification to prevent TCR-mediated graft-versus-host disease (GvHD). Lastly, inclusion of a proprietary "booster molecule" further improves cell expansion, along with phenotype and function, and enables the production of up to hundreds of patient doses from a single manufacturing run.

Significant doses of P-MUC1C-ALLO1 made from multiple healthy donors were achieved and comprised of an exceptionally high-percentage of desirable T_{SCM} cells. Preclinical evaluation of these products showed potent tumor killing and cytokine secretion against MUC1-C-positive breast and ovarian tumor cell lines. P-MUC1C-ALLO1 demonstrates potent cytotoxicity against tumor cells, and minimal killing of normal MUC1-C-positive human primary cells. In a triple-negative breast cancer (TNBC) xenograft model, MUC1C CAR-T eliminated established MDA-MB-468 adenocarcinoma cells, mounted robust T cell expansion in peripheral blood and maintained a favorable T_{SCM} percentage over time. Likewise, in an orthotopic ovarian cancer xenograft model, intraperitoneally administered MUC1C CAR-T eliminated established OVCAR3 adenocarcinoma cells to levels below the limit of detection. All together, these data demonstrated the efficacy of the MUC1C CAR-T cells and the robustness of the allogeneic platform.

P-MUC1C-ALLO1 is an allogeneic T_{SCM} CAR-T therapy that has a potential to treat multiple MUC1 expressing tumor types. P-MUC1C-ALLO1 displayed specificity for tumor vs. normal cells, as well as in vivo efficacy against xenograft models of breast and ovarian cancer. This allogeneic cell therapy is advancing rapidly towards the clinic.

INTRODUCTION

P-MUC1C-ALLO1 Targets Tumor-enriched MUC1-C

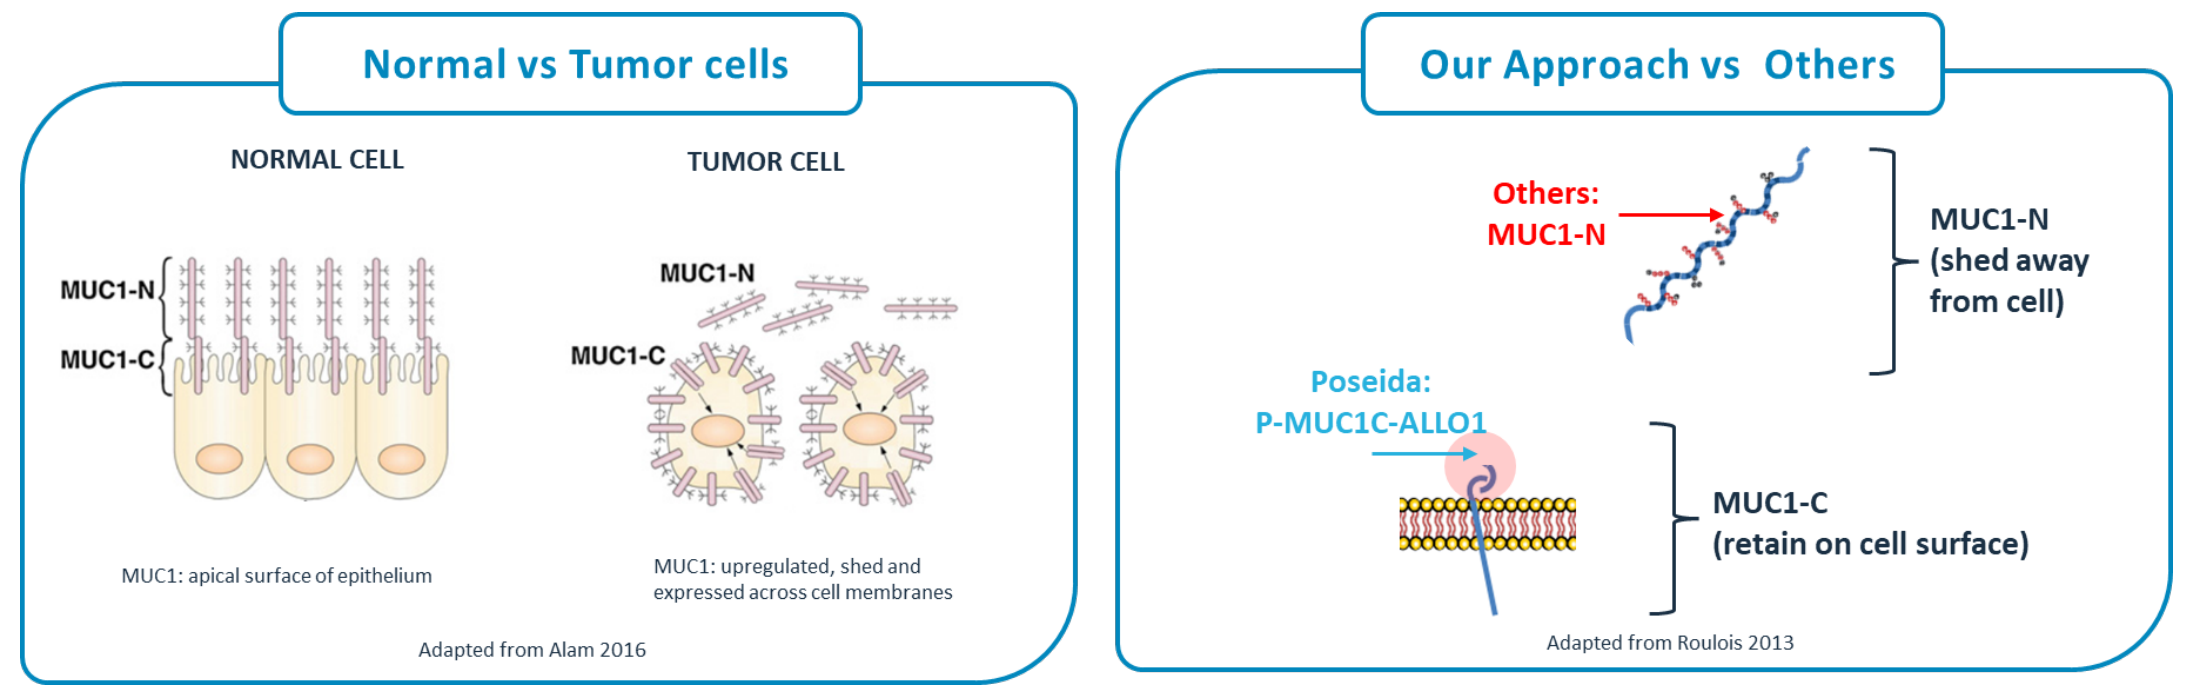

Normal vs Tumor cells
NORMAL CELL
TUMOR CELL
MUC1-N
MUC1-C
MUC1: apical surface of epithelium
MUC1: upregulated, shed and expressed across cell membranes
Adapted from Alam 2016
Our Approach vs Others
Others: MUC1-N
MUC1-N (shed away from cell)
Poseida: P-MUC1C-ALLO1
MUC1-C (retain on cell surface)
Adapted from Roulois 2013

- MUC1 is highly polymorphic and normally expressed on apical surface of epithelium
- On cancer cells, an aberrant form is expressed, and polarity is lost
- P-MUC1C-ALLO1 epitope may be tumor-enriched and is retained on the cell surface following cleavage of MUC1-N

P-MUC1C-ALLO1 has Broad Potential in Solid Tumor

Tumor Type	MUC1-C Expression (% Positive)	N (individual tumor samples assayed)
Stomach	100	5
Endometrium	100	3
Uterine Cervix	100	2
Breast	90	105
Colon	90	62
Ovary	89	135
Lung	80	82
Renal cell	81	42
Oval Cavity	75	4
Rectum	75	4
Vulva	60	5
Esophagus	20	5
Liver	20	5
Skin	0	4
Testis	0	2

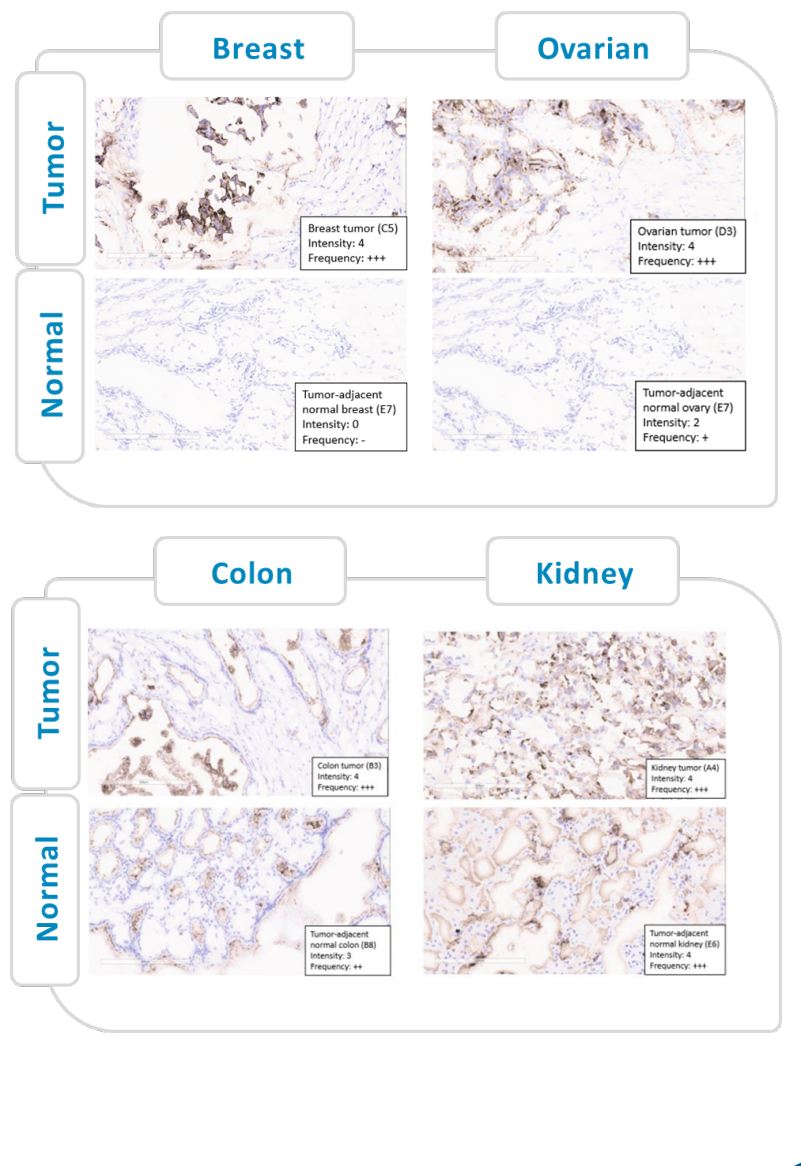

Breast
Ovarian
Tumor
Normal
Colon
Kidney

- IHC staining of frozen human tumor microarray and adjacent normal tissue using MUC1C binder-derived recombinant protein
- P-MUC1C-ALLO1 potentially addresses patient populations in multiple solid tumor indications including many epithelial-derived cancers
- High representation of P-MUC1C-ALLO1 epitope in breast and ovarian cancer

METHODS

P-MUC1C-ALLO1 is a Fully Allogeneic Product

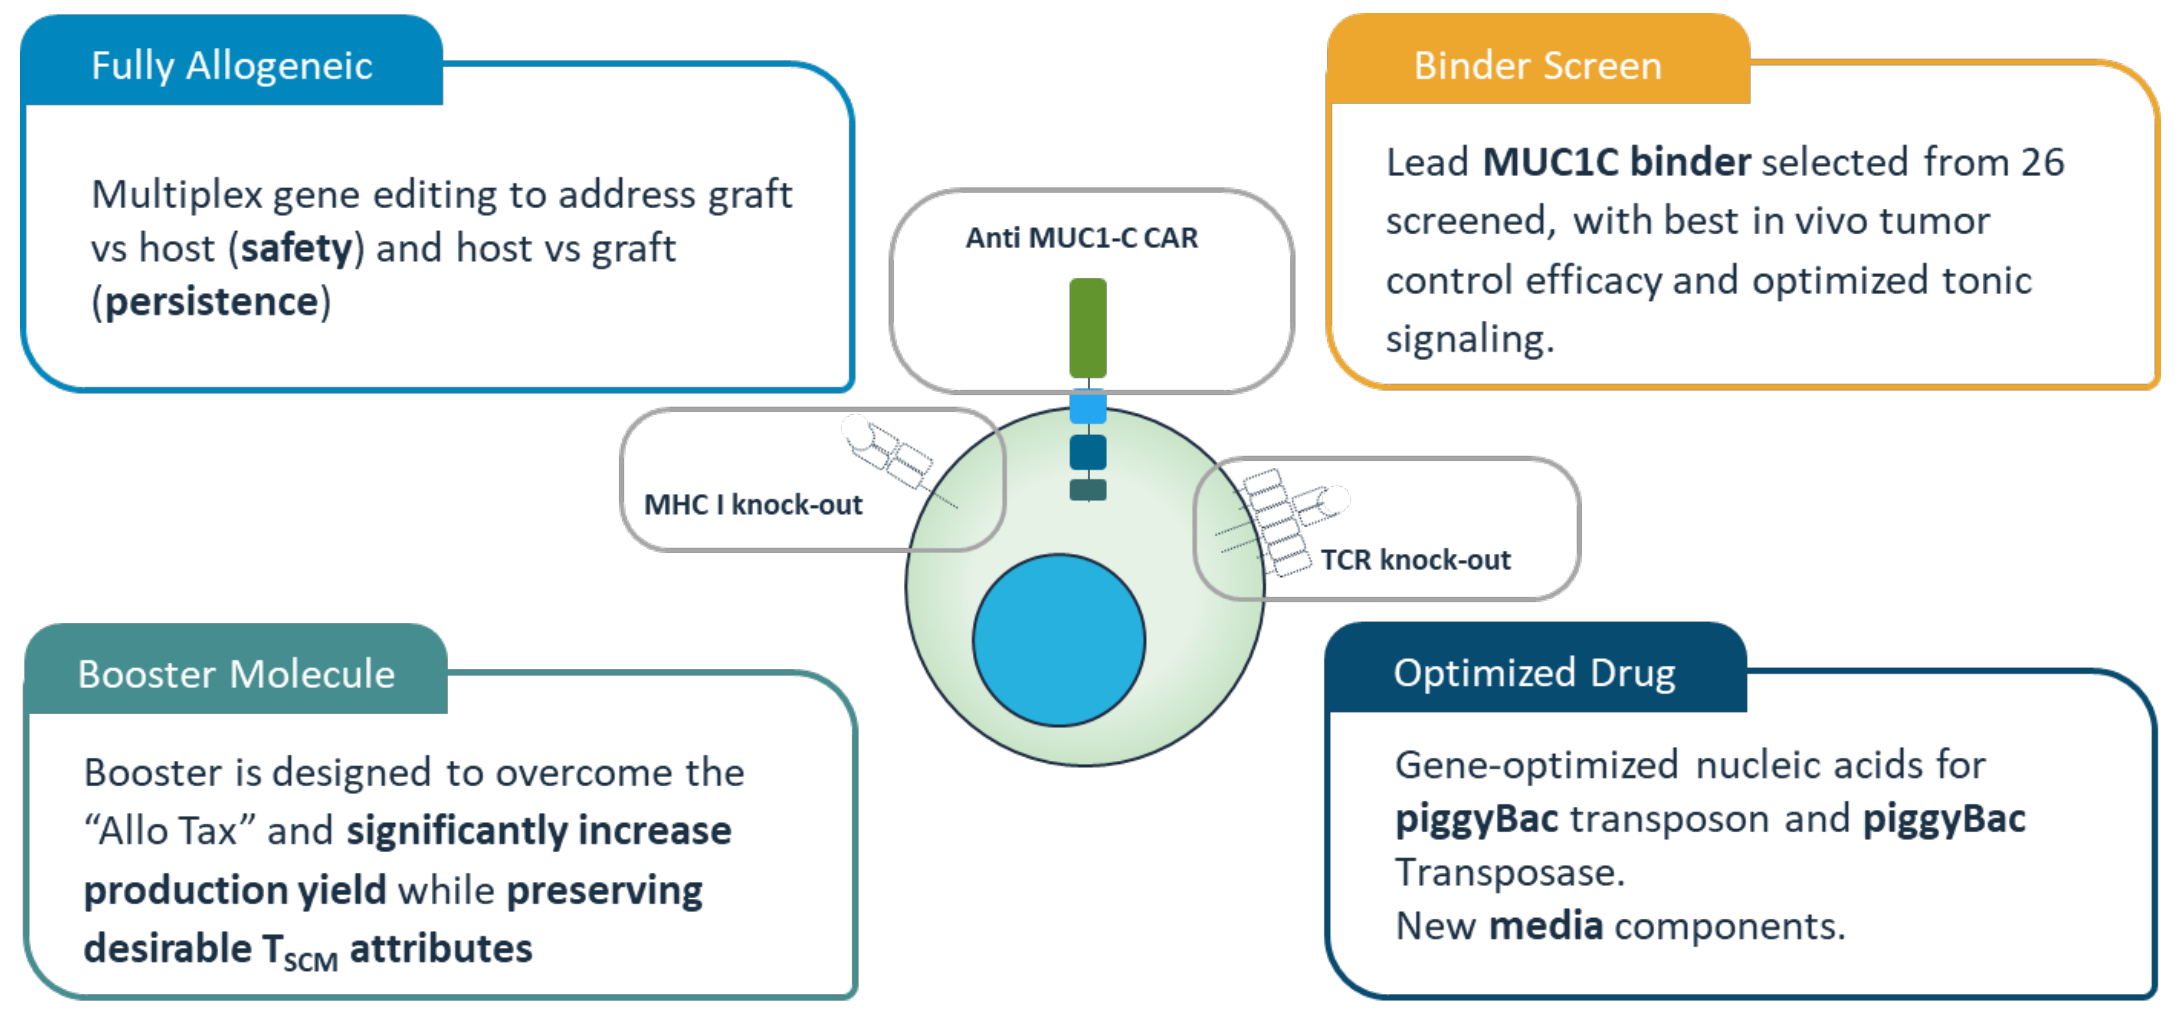

Fully Allogeneic
Multiplex gene editing to address graft vs host (**safety**) and host vs graft (**persistence**)
Binder Screen
Lead **MUC1C binder** selected from 26 screened, with best in vivo tumor control efficacy and optimized tonic signaling.
Anti MUC1-C CAR
MHC I knock-out
TCR knock-out
Booster Molecule
Booster is designed to overcome the "Allo Tax" and **significantly increase production yield** while **preserving desirable T_{SCM} attributes**
Optimized Drug
Gene-optimized nucleic acids for **piggyBac** transposon and **piggyBac** Transposase.
New **media** components.

P-MUC1C-ALLO1 has High T_{SCM} Cell Phenotype

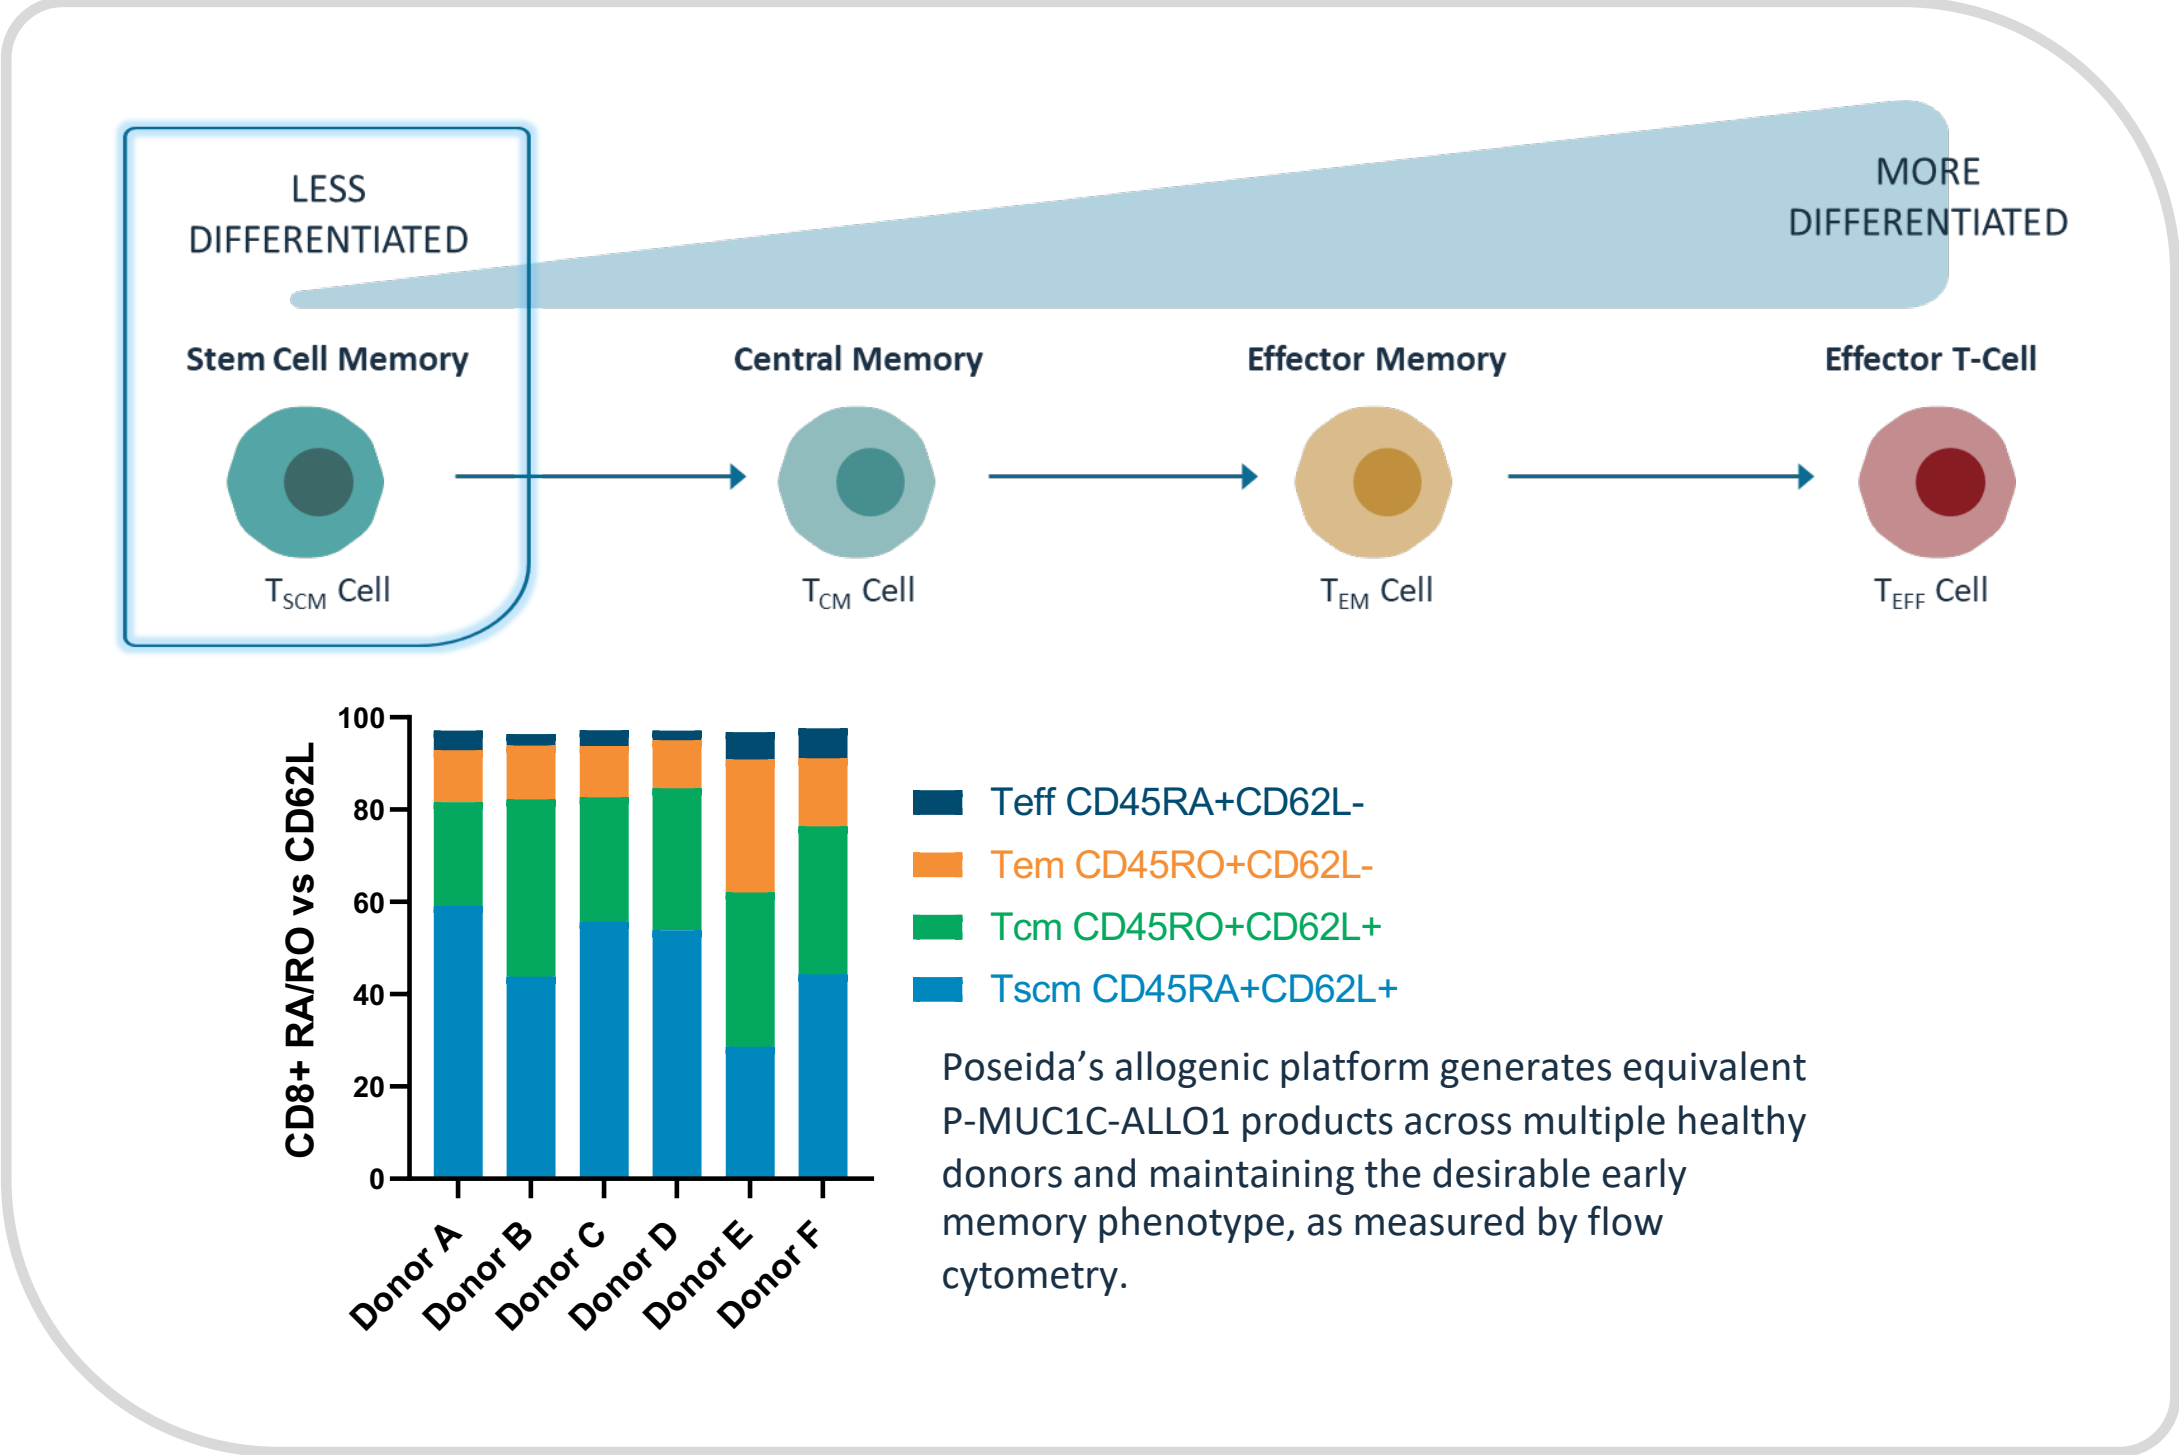

LESS DIFFERENTIATED
MORE DIFFERENTIATED
Stem Cell Memory
Central Memory
Effector Memory
Effector T-Cell
T_{SCM} Cell
T_{CM} Cell
T_{EM} Cell
T_{EFF} Cell
CD8+ RA/RO vs CD62L
Donor A
Donor B
Donor C
Donor D
Donor E
Donor F
Teff CD45RA+CD62L-
Tem CD45RO+CD62L-
Tcm CD45RO+CD62L+
Tscm CD45RA+CD62L+

Poseida's allogenic platform generates equivalent P-MUC1C-ALLO1 products across multiple healthy donors and maintaining the desirable early memory phenotype, as measured by flow cytometry.

P-MUC1C-ALLO1: Manufacturing Process

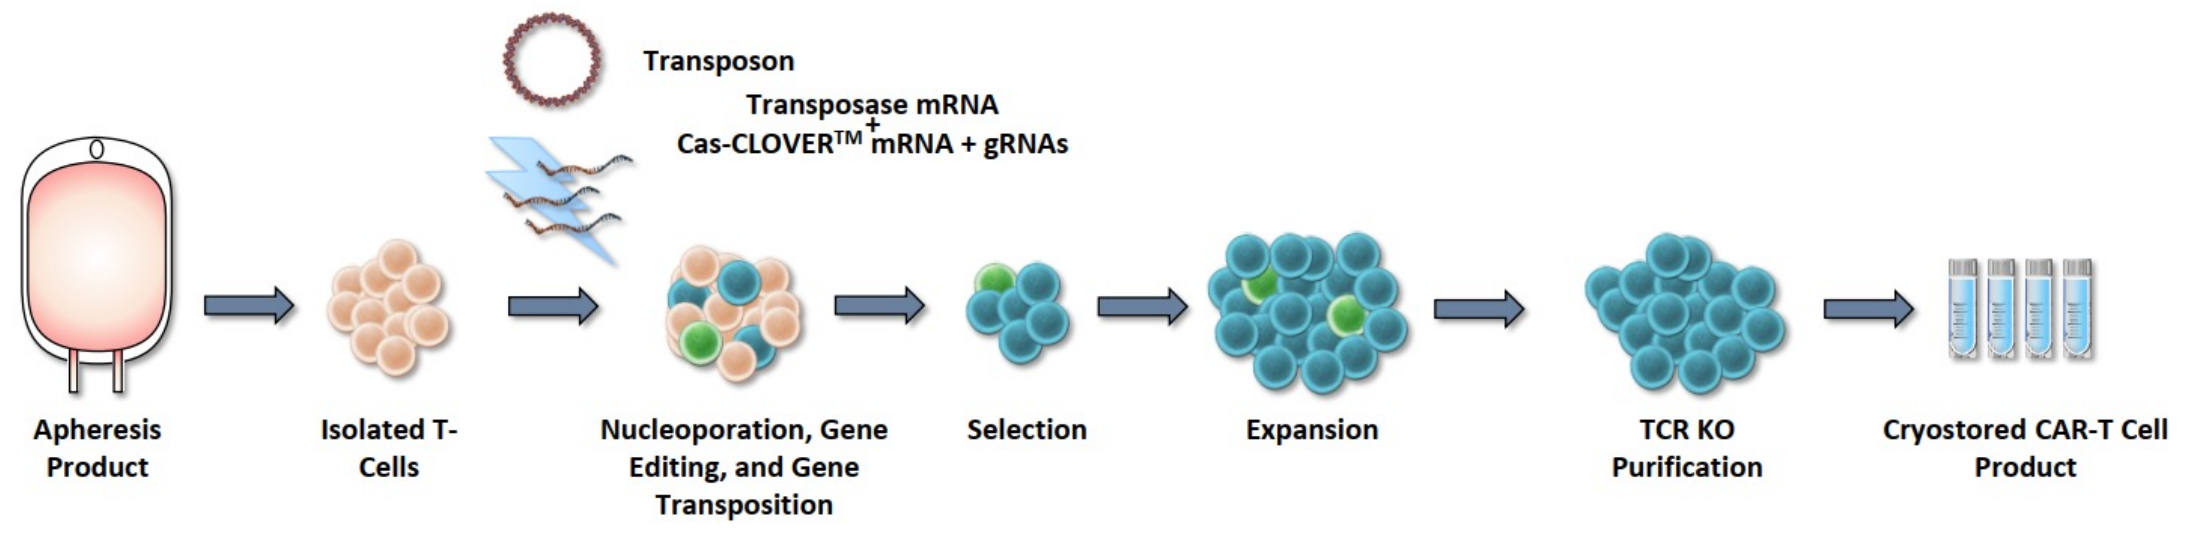

Transposon
Transposase mRNA
+
Cas-CLOVER™ mRNA + gRNAs
Apheresis Product
Isolated T-Cells
Nucleoporation, Gene Editing, and Gene Transposition
Selection
Expansion
TCR KO Purification
Cryostored CAR-T Cell Product

RESULTS

Potent Target-Dependent Cell Killing in vitro

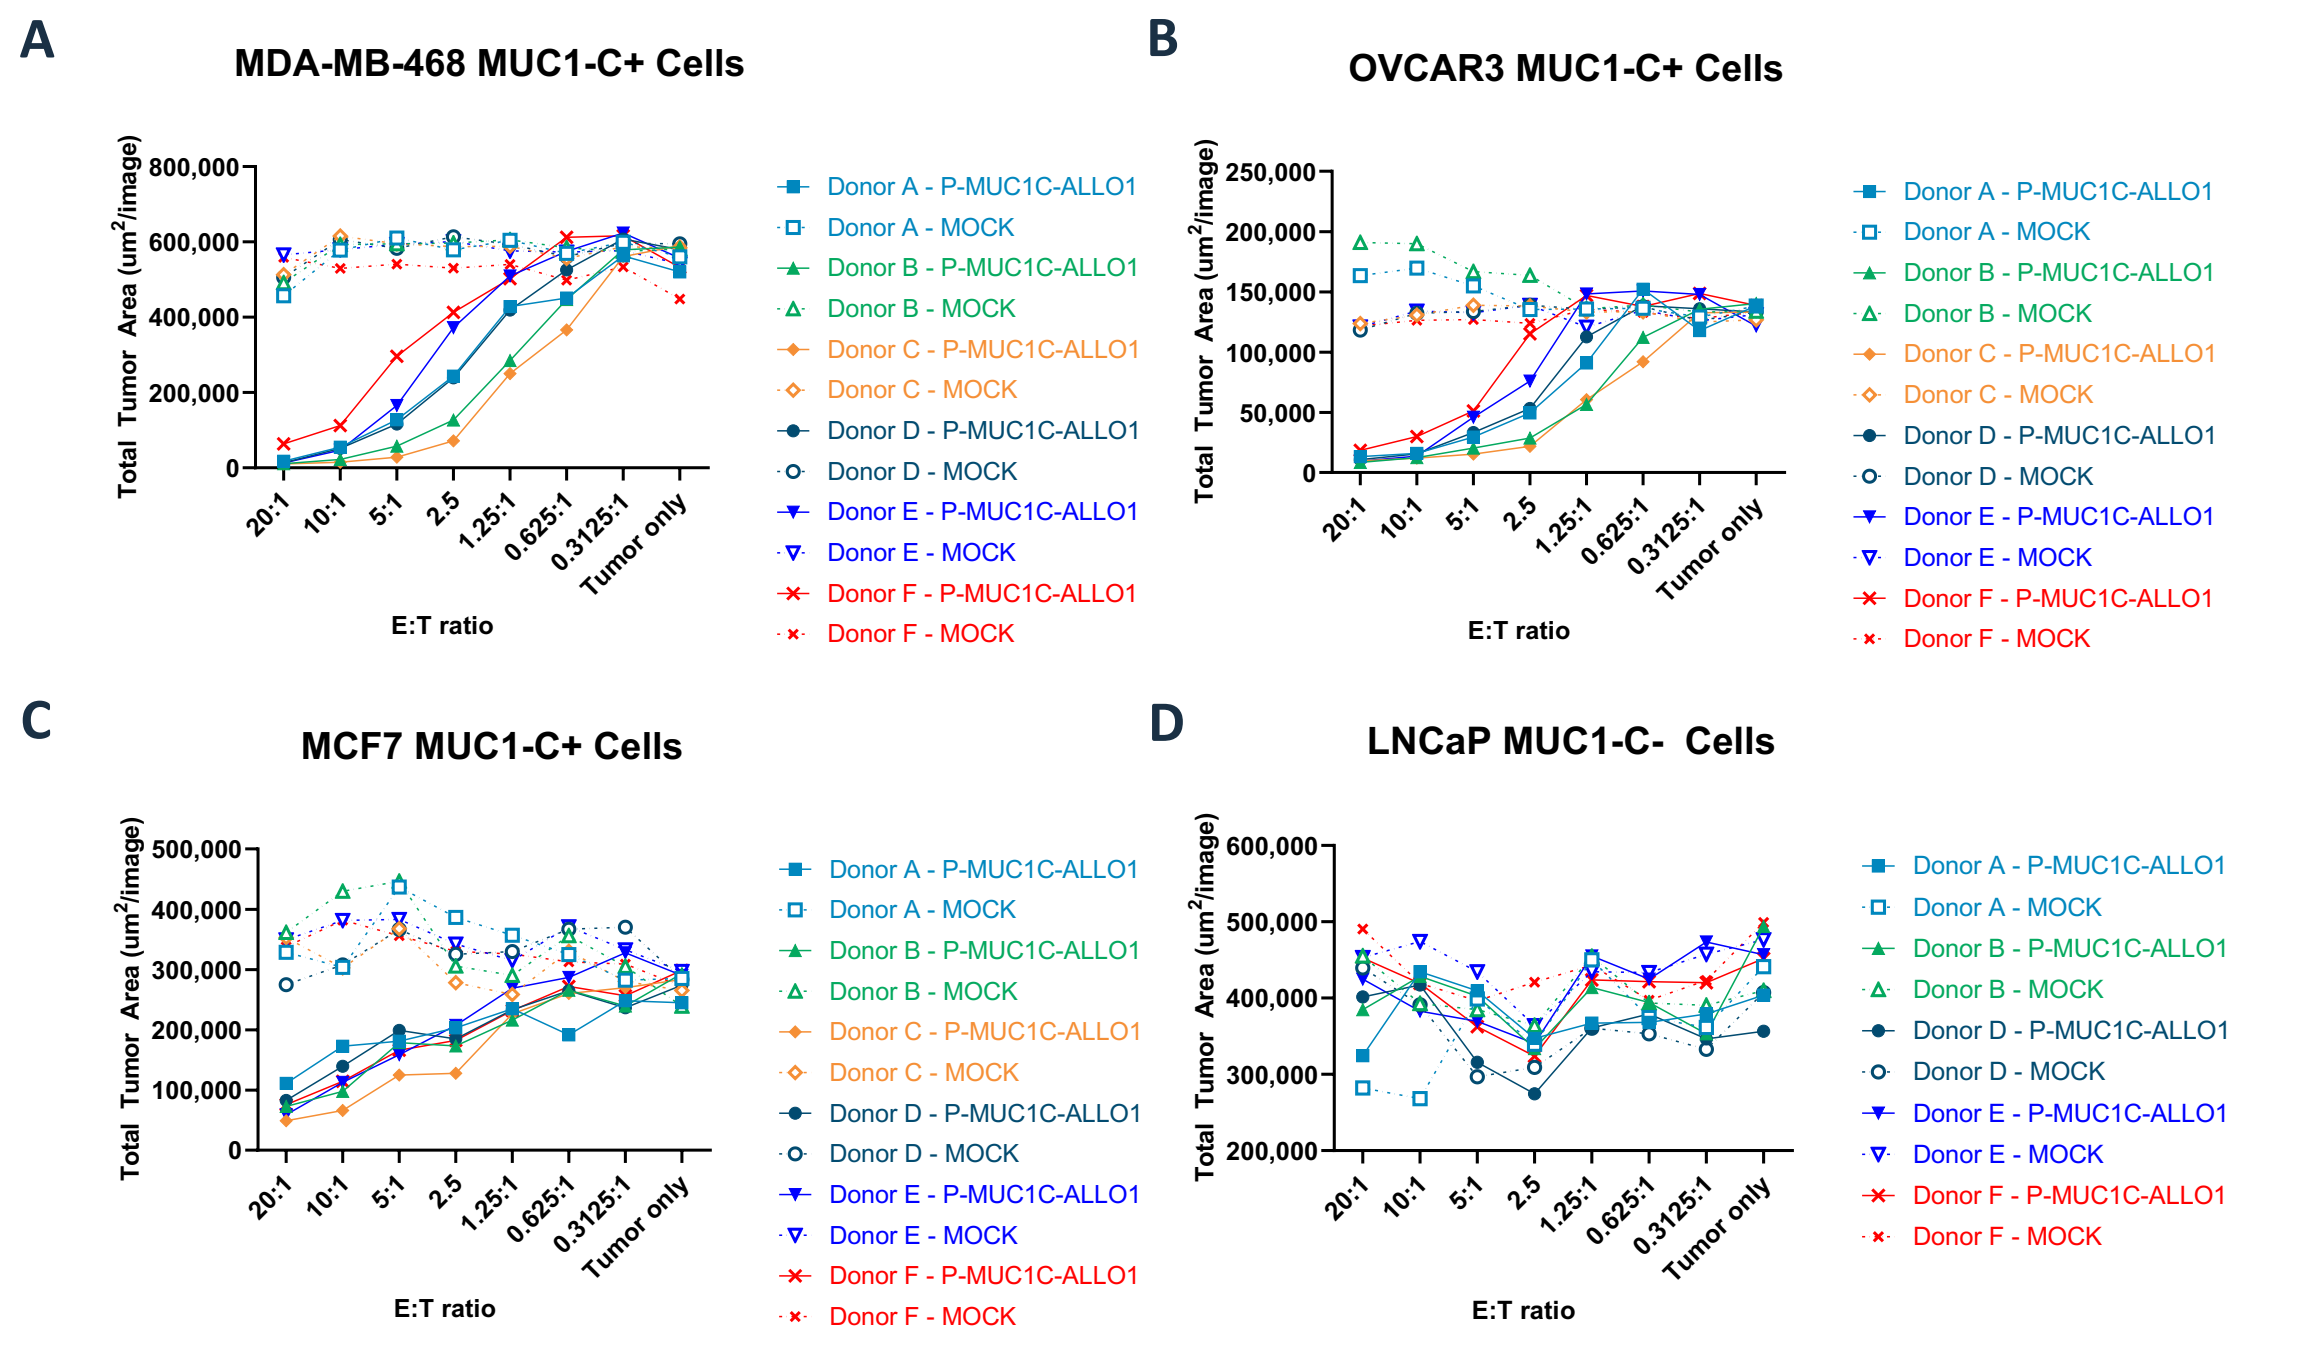

A
MDA-MB-468 MUC1-C+ Cells
B
OVCAR3 MUC1-C+ Cells
C
MCF7 MUC1-C+ Cells
D
LNCaP MUC1-C- Cells
Total Tumor Area (um²/image)
E:T ratio
Donor A - P-MUC1C-ALLO1
Donor A - MOCK
Donor B - P-MUC1C-ALLO1
Donor B - MOCK
Donor C - P-MUC1C-ALLO1
Donor C - MOCK
Donor D - P-MUC1C-ALLO1
Donor D - MOCK
Donor E - P-MUC1C-ALLO1
Donor E - MOCK
Donor F - P-MUC1C-ALLO1
Donor F - MOCK

Figure 1. Specific killing of P-MUC1C-ALLO1 CAR-T cells against MUC1-C positive tumor cells, but not MUC1-C negative tumor cell. Cytolytic activity CAR-T against MUC1-C positive TNBC cell MDA-MB-468.GFP (A), ovarian cancer cell OVCAR3.GFP (B), breast cancer cell MCF7.GFP (C) and MUC1-C negative prostate cancer cell LNCaP.GFP (D) at different effector to target (E:T) ratio was measured using Incucyte after co-culture for 48 hours ~ 62 hours.

Robust anti-Tumor Efficacy in TNBC Model

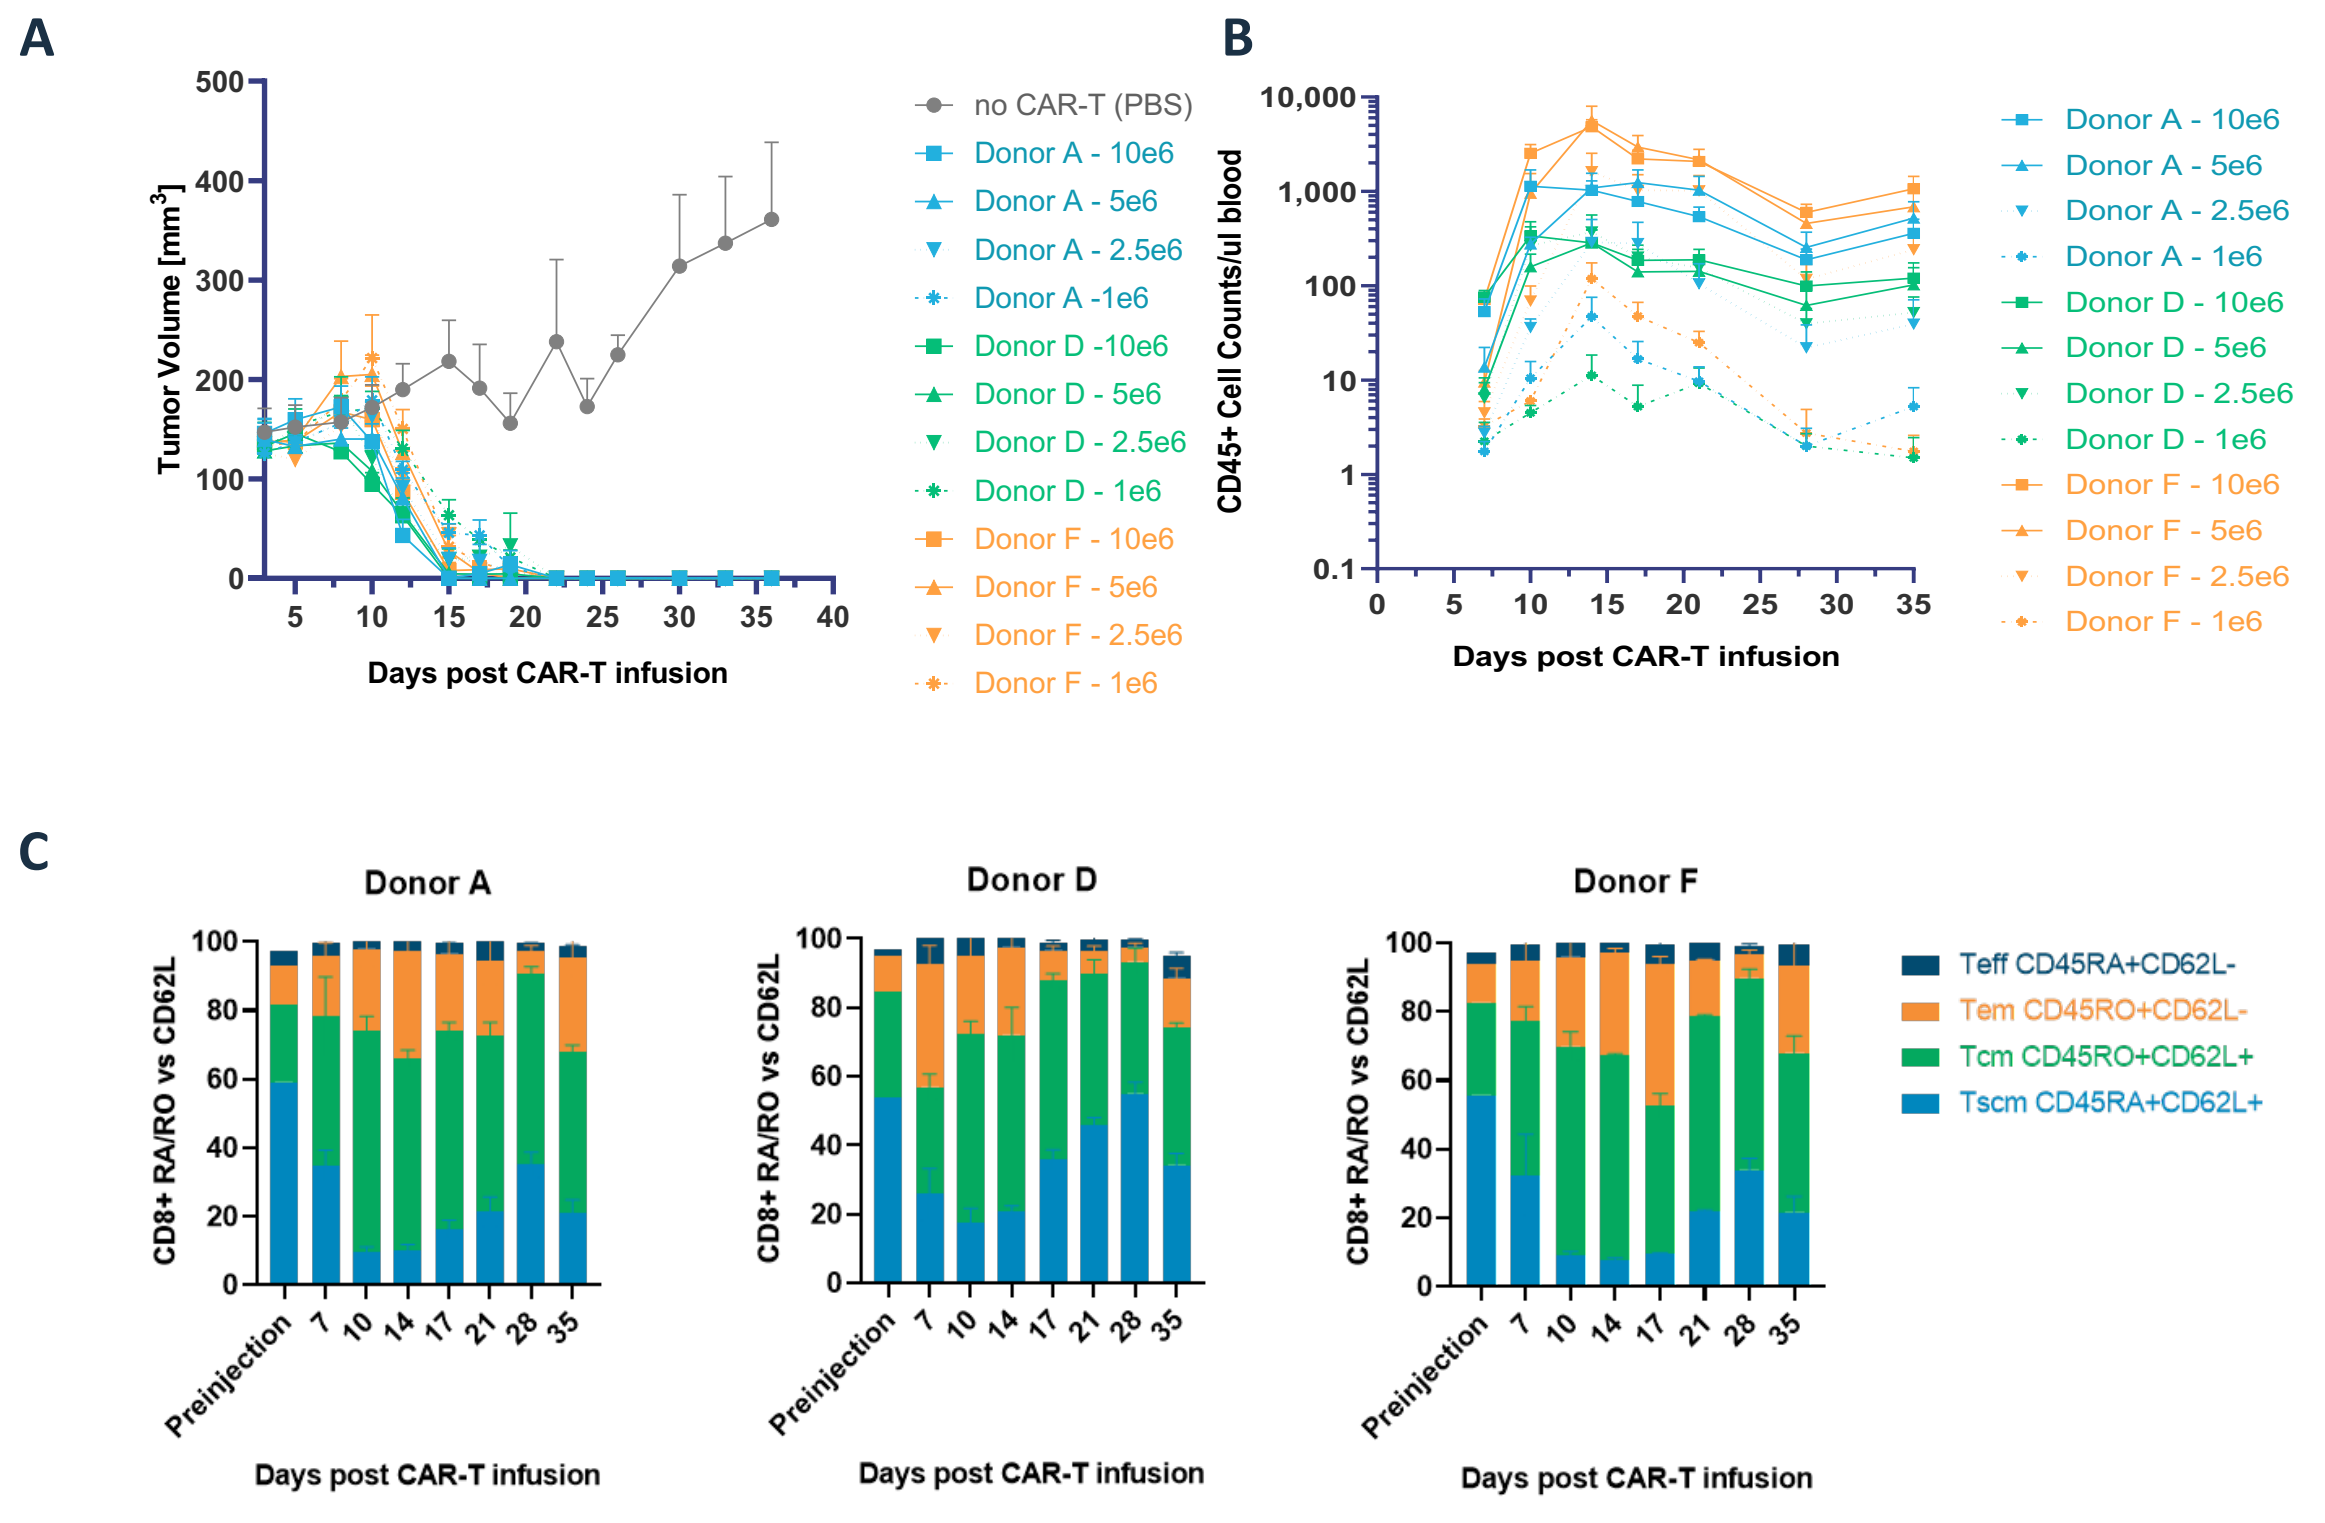

A
Tumor Volume [mm³]
Days post CAR-T infusion
no CAR-T (PBS)
Donor A - 10e6
Donor A - 5e6
Donor A - 2.5e6
Donor A -1e6
Donor D -10e6
Donor D - 5e6
Donor D - 2.5e6
Donor D - 1e6
Donor F - 10e6
Donor F - 5e6
Donor F - 2.5e6
Donor F - 1e6
B
CD45+ Cell Counts/ul blood
C
Donor A
Donor D
Donor F
CD8+ RA/RO vs CD62L
Preinjection
Teff CD45RA+CD62L-
Tem CD45RO+CD62L-
Tcm CD45RO+CD62L+
Tscm CD45RA+CD62L+

Figure 2. P-MUC1C-ALLO1 cells demonstrate robust efficacy in TNBC MDA-MB-468 xenograft models. Caliper measurements of NSG mice bearing established subcutaneous MDA-MB-468 TNBC tumors. Mice were treated intraperitoneally with the indicated doses of MUC1C CAR-T cells at day 0. Tumor volume assessments by caliper measurement across study timepoints are shown (A). P-MUC1C-ALLO1 CAR-T proliferates in peripheral Blood. Total CAR-T cells in blood as measured at study time points by TruCount staining for human CD45+ cells per µl for all animals (B). Phenotype of CD8+ T cells in blood as measured by FACS staining for all animals as displayed as group averages at 10^6 CAR-T dose with error bars as SEM (C).

RESULTS

Robust anti-Tumor Efficacy in Ovarian Cancer Model

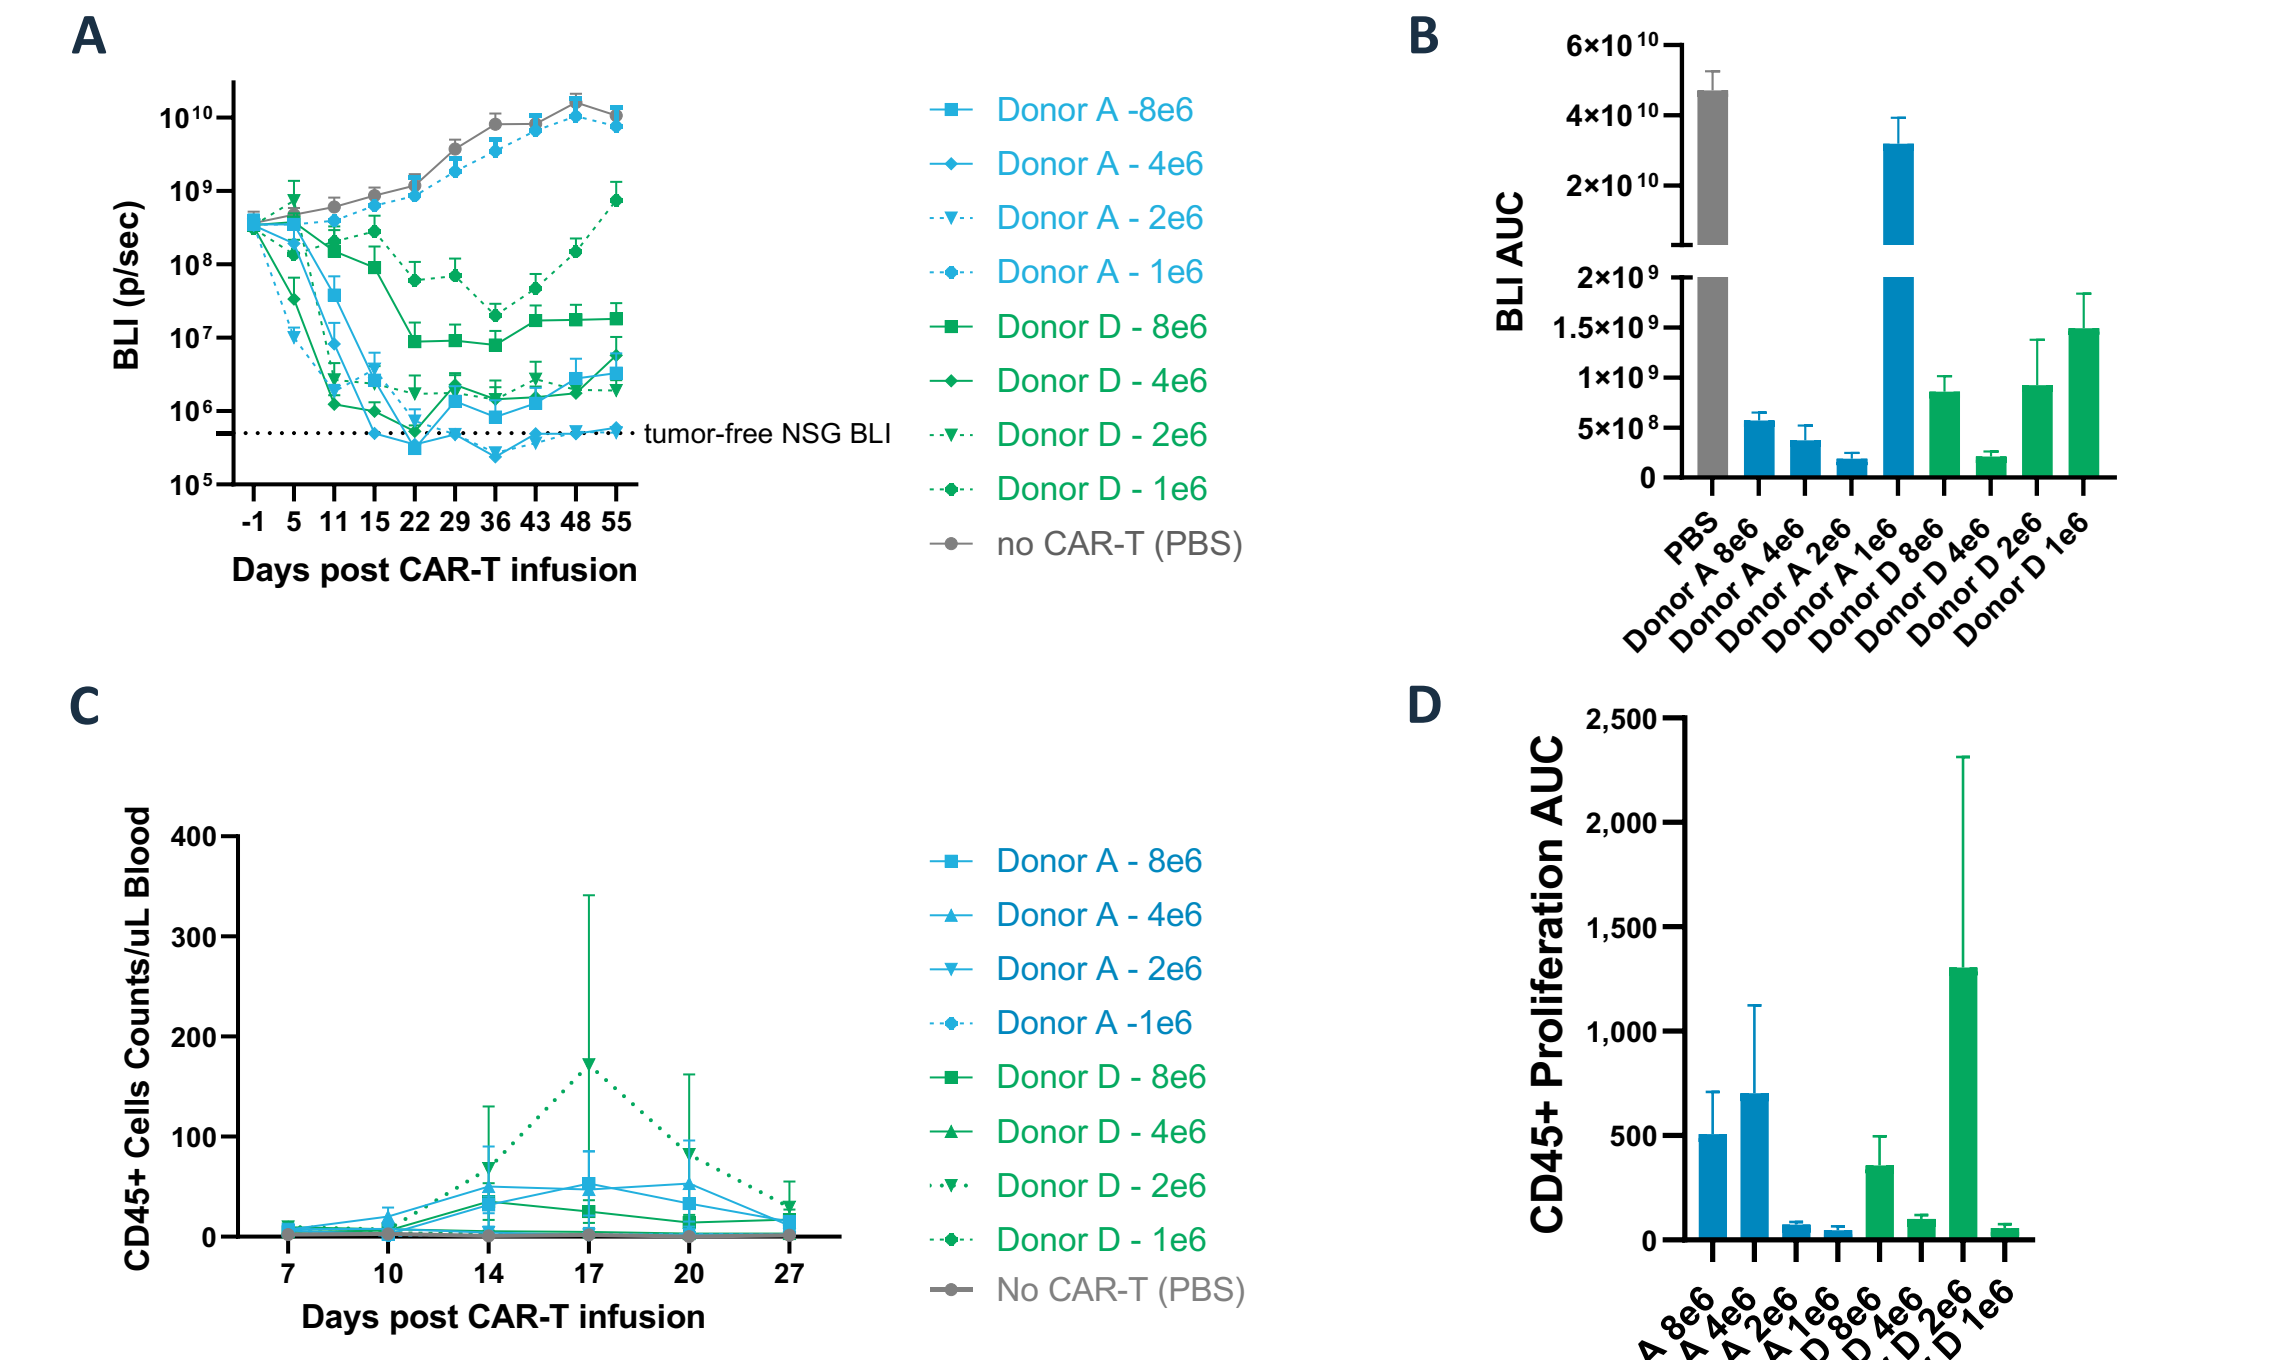

A
BLI (p/sec)
tumor-free NSG BLI
Days post CAR-T infusion
Donor A -8e6
Donor A - 4e6
Donor A - 2e6
Donor A - 1e6
Donor D - 8e6
Donor D - 4e6
Donor D - 2e6
Donor D - 1e6
no CAR-T (PBS)
B
BLI AUC
PBS
C
CD45+ Cells Counts/uL Blood
No CAR-T (PBS)
D
CD45+ Proliferation AUC

Figure 3. P-MUC1C-ALLO1 cells demonstrate robust efficacy in ovarian cancer OVCAR3 xenograft models. Body Luminescence Intensity (BLI) measurements of NSG mice bearing established intraperitoneal OVCAR3 tumors. Mice were treated intraperitoneally with the indicated doses of MUC1C CAR-T cells at day 0. Tumor volume assessments by BLI measurement across study timepoints are shown (A). Area under the curve with SEM (Standard Error of the Mean) for tumor sizes is shown for each treated group in the bar graph (B). P-MUC1C-ALLO1 CAR-T could traffic to peripheral Blood. Total CAR-T cells in blood as measured at study time points by TruCount staining for human CD45+ cells per µl for all animals (C). Area under the curve with SEM (Standard Error of the Mean) for T cell number is shown for each treated group in the bar graph (D).

Excellent Tumor vs. Normal Cell Specificity

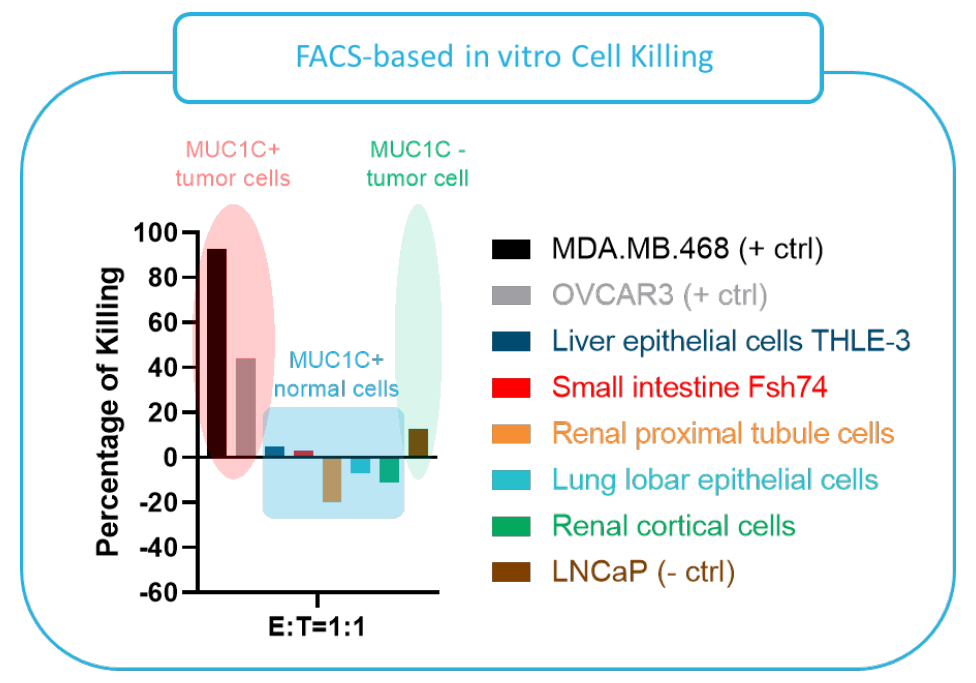

FACS-based in vitro Cell Killing
MUC1C+ tumor cells
MUC1C - tumor cell
MUC1C+ normal cells
Percentage of Killing
E:T=1:1
MDA.MB.468 (+ ctrl)
OVCAR3 (+ ctrl)
Liver epithelial cells THLE-3
Small intestine Fsh74
Renal proximal tubule cells
Lung lobar epithelial cells
Renal cortical cells
LNCaP (- ctrl)

Figure 4. P-MUC1C-ALLO1 demonstrates excellent cytotoxicity against MUC1-C positive tumor cells, and minimal killing of MUC1-C-positive human normal primary cells. While MUC1C CAR-T cells demonstrates 40% to 90% of target cell killing against MUC1-C+ tumor cells (MDA-MB-468 and OVCAR3), P-MUC1C-ALLO1 has minimal cytotoxicity against MUC1-C+ normal cells after 48 hours coculture at 1:1 E:T Ratio. Prostate cancer cell line LNCaP has minimal MUC1-C+ expression and is serving as negative control. End-point FACS analysis was used to measured cytotoxicity. Cytotoxicity was represented as the percent of targets killed and was calculated as the average number of target cells remaining after coculture with P-MUC1C-ALLO1 CAR-T cells relative to cocultures with Mock T-cell controls.

CONCLUSIONS

- P-MUC1C-ALLO1 is Poseida's allogeneic CAR T_{SCM} product that has a potential to treat multiple MUC1 expressing tumor types.
- P-MUC1C-ALLO1 has potent in vivo efficacy against xenograft models of breast and ovarian cancer.
- P-MUC1C-ALLO1 displayed specificity for tumor vs. normal cells.

REFERENCES

- Alam, Maroof, et al. "MUC1-C represses the Crumbs complex polarity factor CRB3 and downregulates the Hippo pathway." Molecular Cancer Research 14.12 (2016): 1266-1276.
- Roulois, David, Marc Grégoire, and Jean-François Fonteneau. "MUC1-specific cytotoxic T lymphocytes in cancer therapy: induction and challenge." BioMed research international 2013 (2013).