P-BCMA-ALLO1 — A Fully Allogeneic Stem Cell Memory T Cell (T_{SCM}) CAR-T Therapy Targeting BCMA for the Treatment of Multiple Myeloma Shows Potent Anti-Tumor Activity

Maximilian Richter, Stacey A. Cranert, Deepak Patil, Yan Zhang, Yening Tan, Min Tong, Christine Domingo, Leslie Weiss, Karl Marquez, Jessica Sparks, Elvira Argus, Samad A. Ibitokou, Christopher E. Martin, Eric M. Ostertag, Sumiti Jain, Julia Coronella, Devon J. Shedlock

Poseida Therapeutics, Inc., 9390 Towne Centre Drive, Suite 200, San Diego, CA, 92121

POSEIDA THERAPEUTICS

ABSTRACT

Emerging data from the clinic highlight a great potential of autologous CAR-T cell therapies in the treatment of refractory/relapsed multiple myeloma (MM). Despite their promise, autologous CAR-T cell therapies face many manufacturing challenges including consistency, product variability, toxicity and cost. However, the production of allogeneic off-the-shelf CAR-T cell therapies from healthy donor T cells can address these concerns.

Using Poseida's proprietary non-viral piggyBac® (PB) DNA Delivery System, in combination with the high-fidelity Cas-CLOVER™ (CC) Site-Specific Gene Editing System, we have engineered a fully allogeneic P-BCMA-ALLO1 product candidate for MM. P-BCMA-ALLO1 is generated by editing healthy donor T cells using CC to eliminate surface expression of both TCR and MHC class I, with little to no off-target activity for optimal safety. Moreover, CC efficiently edits resting T cells that have not yet undergone activation, a critical element of our process that uniquely yields a T_{SCM} enriched allogeneic CAR-T product candidate. T_{SCM} have been correlated with best responses and superior safety in the clinical trial of our P-BCMA-101 autologous product candidate. Inclusion of a "booster molecule" in our allogeneic manufacturing process further improves the expansion of gene-edited cells and enables the production of hundreds of patient doses from a single manufacturing run, thereby reducing the manufacturing cost per dose into the same range as that of a monoclonal antibody. In addition to the CAR, the PB system enables the delivery of a safety switch, which can be used to rapidly deplete some or all of the administered cells in case of an adverse event, as well as a selectable marker allowing the generation of a final cell product that is >95% CAR-positive. In addition to selective CAR+ enrichment during expansion, our final product also undergoes depletion of residual CD3+/TCR+ cells in a robust clinical scale manufacturing process, resulting in a safe TSCM enriched allogeneic BCMA CAR-T product for MM.

Preclinical in vitro evaluation of P-BCMA-ALLO1 showed effective target-dependent killing, cytokine secretion as well as high proliferative capacity in serial restimulation assays, as we expect from Poseida's unique high percentage T_{SCM} product candidates. Importantly, the gene-edited allogeneic product exhibited similar or superior performance compared to an autologous product, both in vitro and in multiple in vivo immunodeficient mouse (NSG) models. In an aggressive systemic MM.1S model, P-BCMA-ALLO1 from multiple donors induced long-term tumor control after a single dose administration. Notably, anti-tumor efficacy and in vivo CAR-T expansion observed for P-BCMA-ALLO1 was comparable to that of non-gene edited healthy donor CAR-T cell controls.

The P-BCMA-ALLO1 product is generated using multiple proprietary components of Poseida's technology that aim to confer efficacy and persistence, safety, and cost effective and consistent manufacturing of our product candidate in the clinic. P-BCMA-ALLO1 could potentially make a significant impact on the treatment of MM.

INTRODUCTION

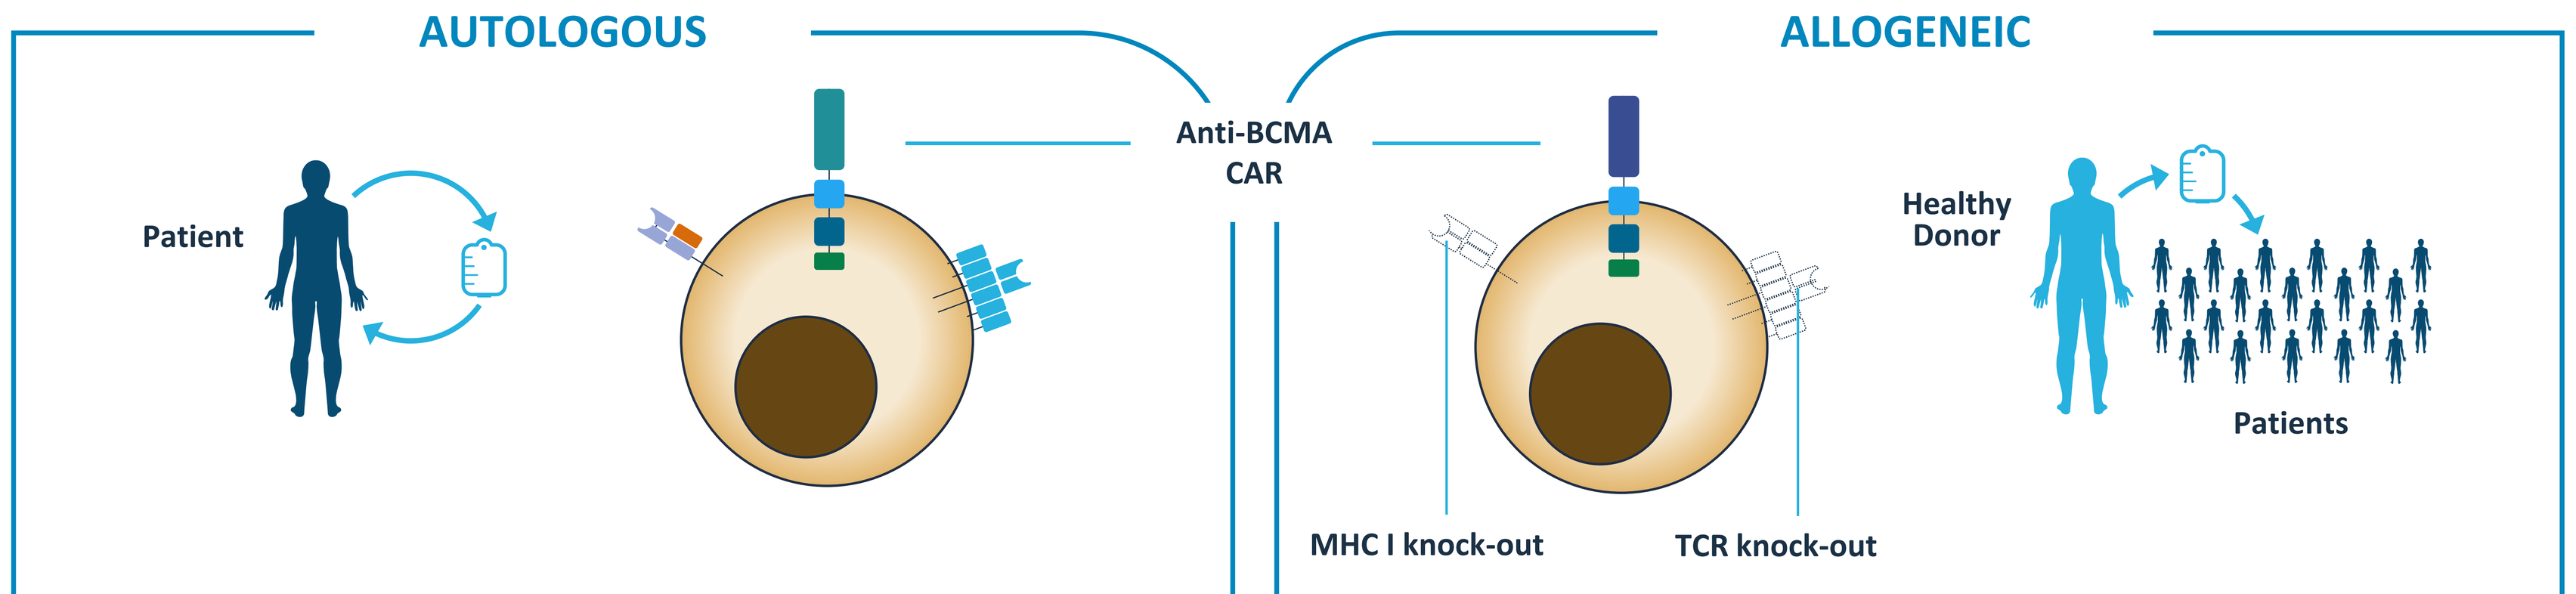
AUTOLOGOUS
ALLOGENEIC
Patient
Anti-BCMA CAR
Healthy Donor
Patients
MHC I knock-out
TCR knock-out

CHALLENGES
- **Variable manufacturability** of patient material
- Long manufacturing **time**
- High **cost** of an individualized product

ADVANTAGES
- **Quality:** Healthy donor material for consistent potency, higher $\%T_{SCM}$ and ease of manufacturing
- **Universal:** Off-the shelf product for increased patient access
- **Scalability/cost:** Large number of doses for numerous patients from a single manufacturing run

P-BCMA-ALLO1 Manufacturing Process

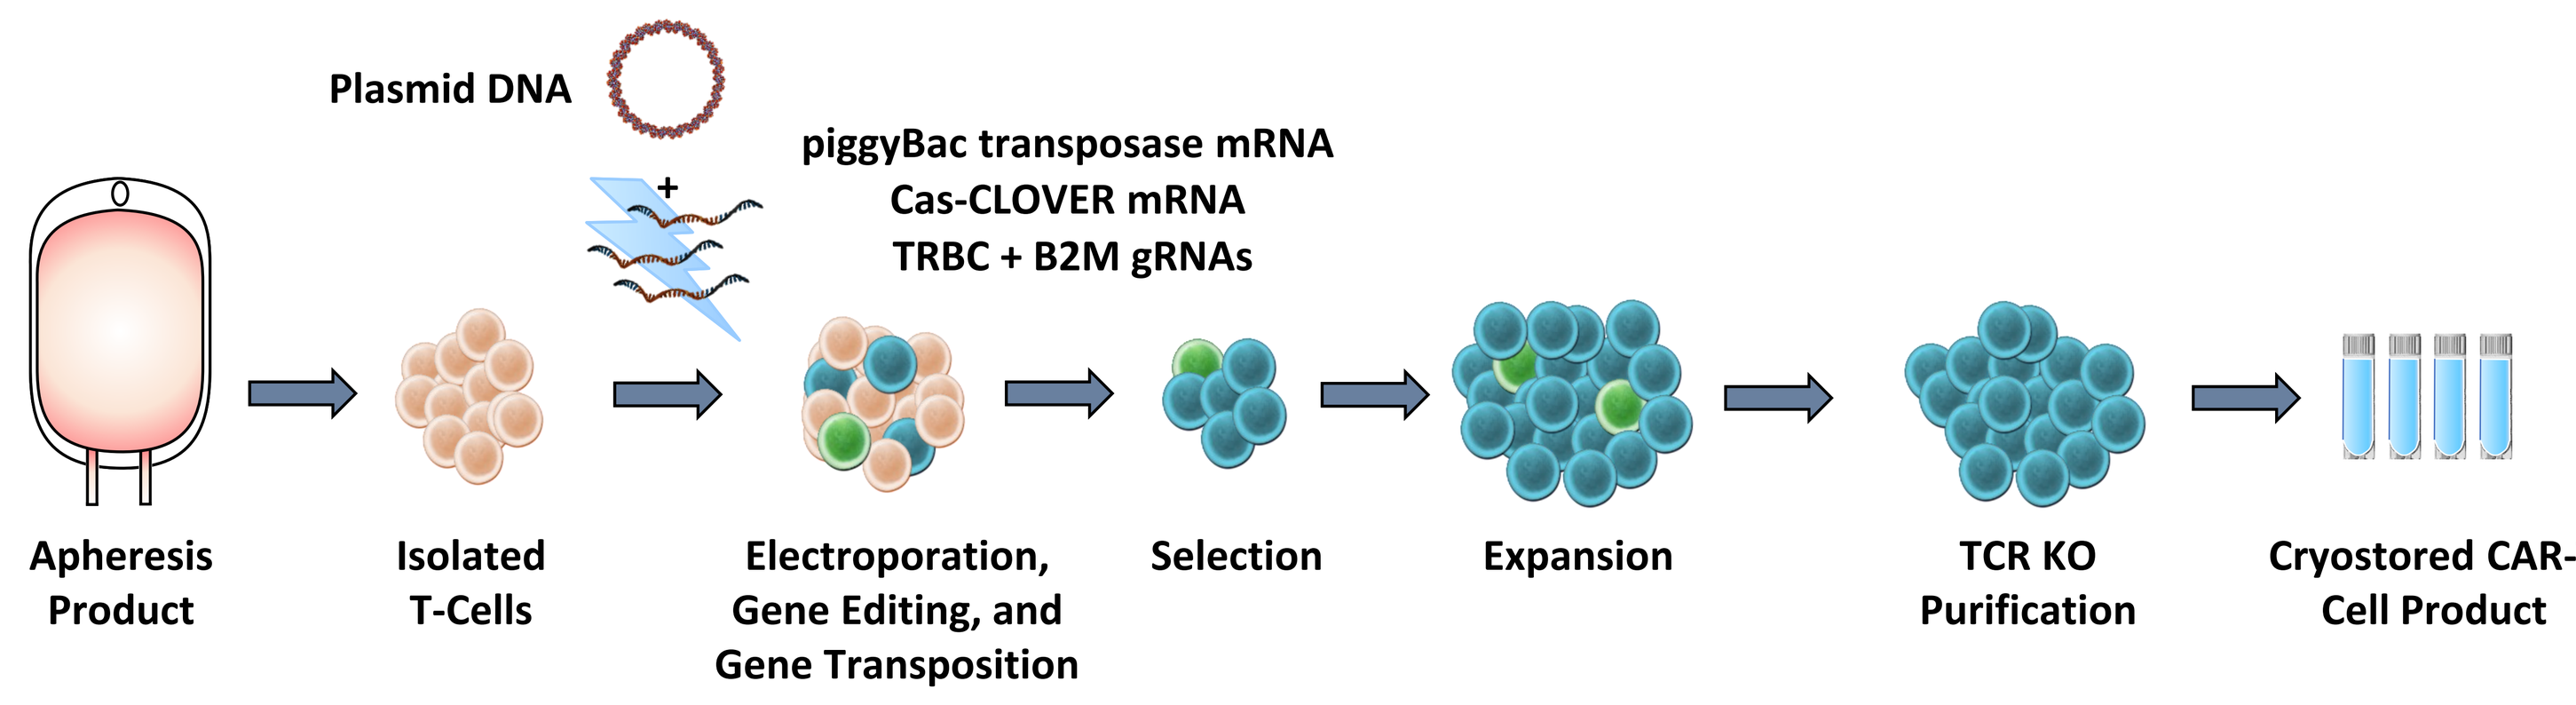
Plasmid DNA
piggyBac transposase mRNA
Cas-CLOVER mRNA
TRBC + B2M gRNAs
Apheresis Product
Isolated T-Cells
Electroporation, Gene Editing, and Gene Transposition
Selection
Expansion
TCR KO Purification
Cryostored CAR-T Cell Product

Fig. 1 P-BCMA-ALLO1 manufacturing using the non-viral piggyBac DNA delivery system and the Cas-CLOVER gene editing system yields a fully allogeneic, off-the-shelf anti-BCMA CAR-T. Healthy donor T cells are isolated from apheresis product and electroporated to deliver a transposon plasmid DNA, piggyBac mRNA, Cas-CLOVER mRNA as well as guide RNAs targeting TCR and MHC class I. The delivered transposon encodes the anti-BCMA CAR, an inducible safety switch as well as a selectable marker. Successfully transposed CAR-T cells are then selected and expanded. At the end of production, residual TCR-positive T cells are depleted, and the final CAR-T cell product is cryopreserved.

P-BCMA-ALLO1 is Rich in T Stem Cell Memory Cells

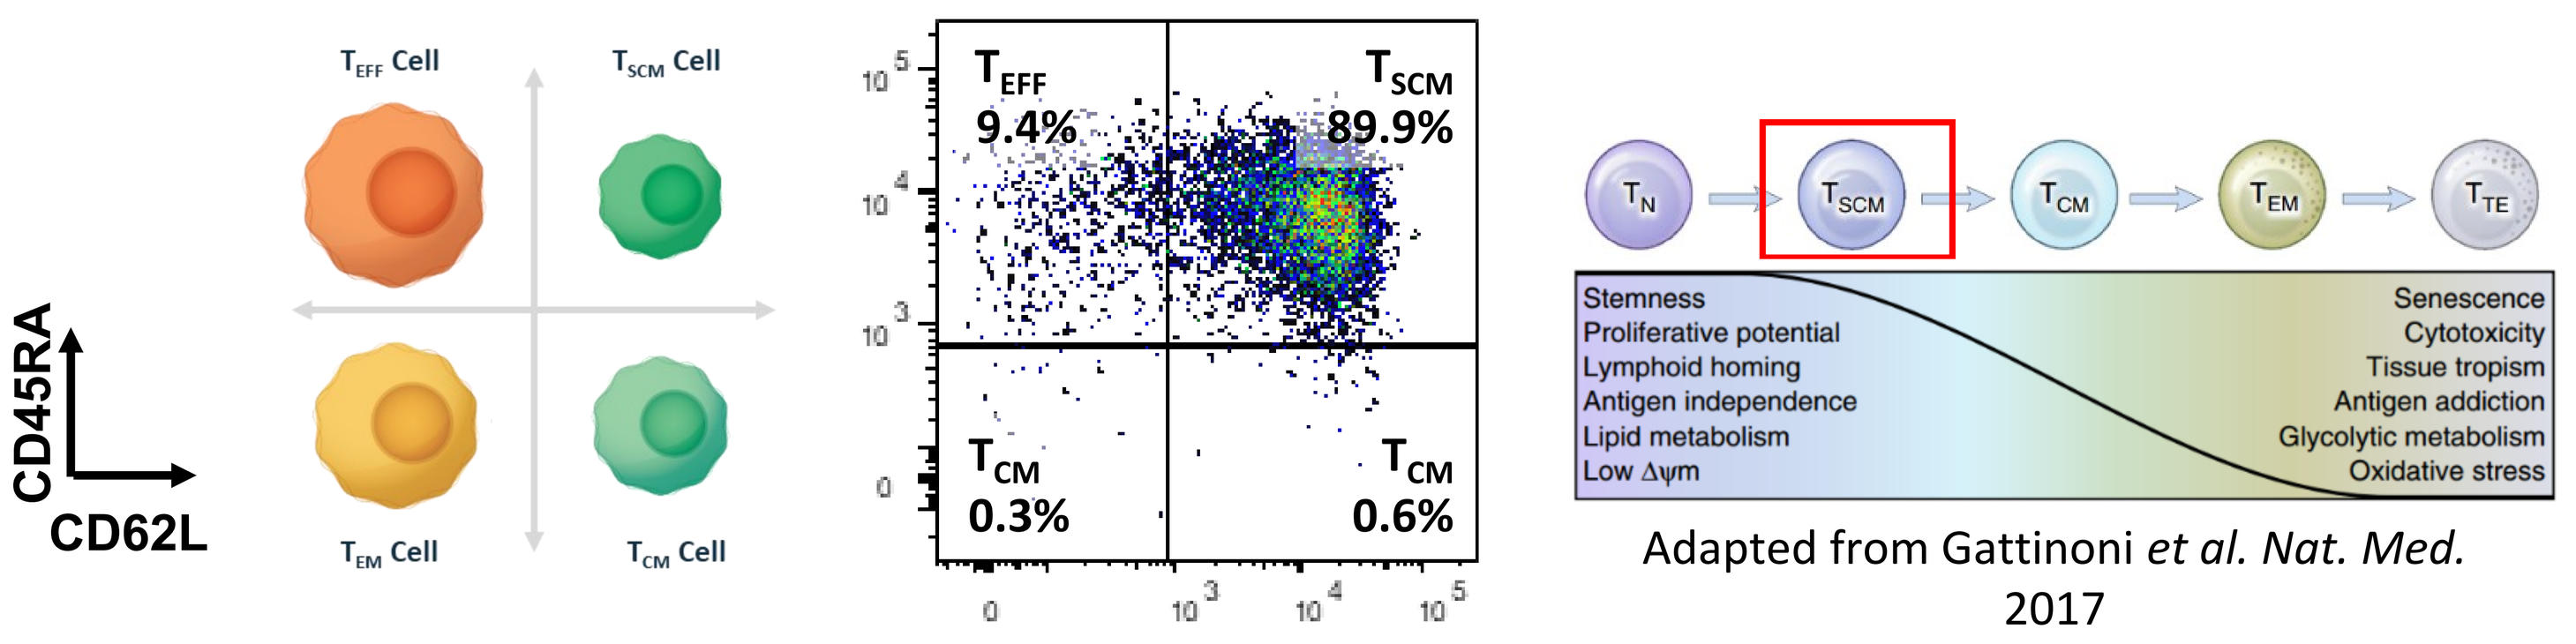
CD45RA
CD62L
T_{EFF} Cell
T_{SCM} Cell
T_{EM} Cell
T_{CM} Cell
T_{EFF} 9.4%
T_{SCM} 89.9%
T_{CM} 0.3%
T_{CM} 0.6%
Stemness
Proliferative potential
Lymphoid homing
Antigen independence
Lipid metabolism
Low Δψm
Senescence
Cytotoxicity
Tissue tropism
Antigen addiction
Glycolytic metabolism
Oxidative stress
Adapted from Gattinoni *et al. Nat. Med.* 2017

Fig. 2 P-BCMA-ALLO1 is rich in T stem cell memory cells (T_{SCM}). PiggyBac gene delivery and Cas-CLOVER gene editing in resting T cells allows for the generation of an allogeneic CAR-T product rich in T_{SCM}. These cells are long-lived, self-renewing, and multipotent, making them a desirable T cell population for adoptive immunotherapy applications.

METHODS & RESULTS

Cas-CLOVER Gene Editing for Efficient TCR KO in Resting T Cells

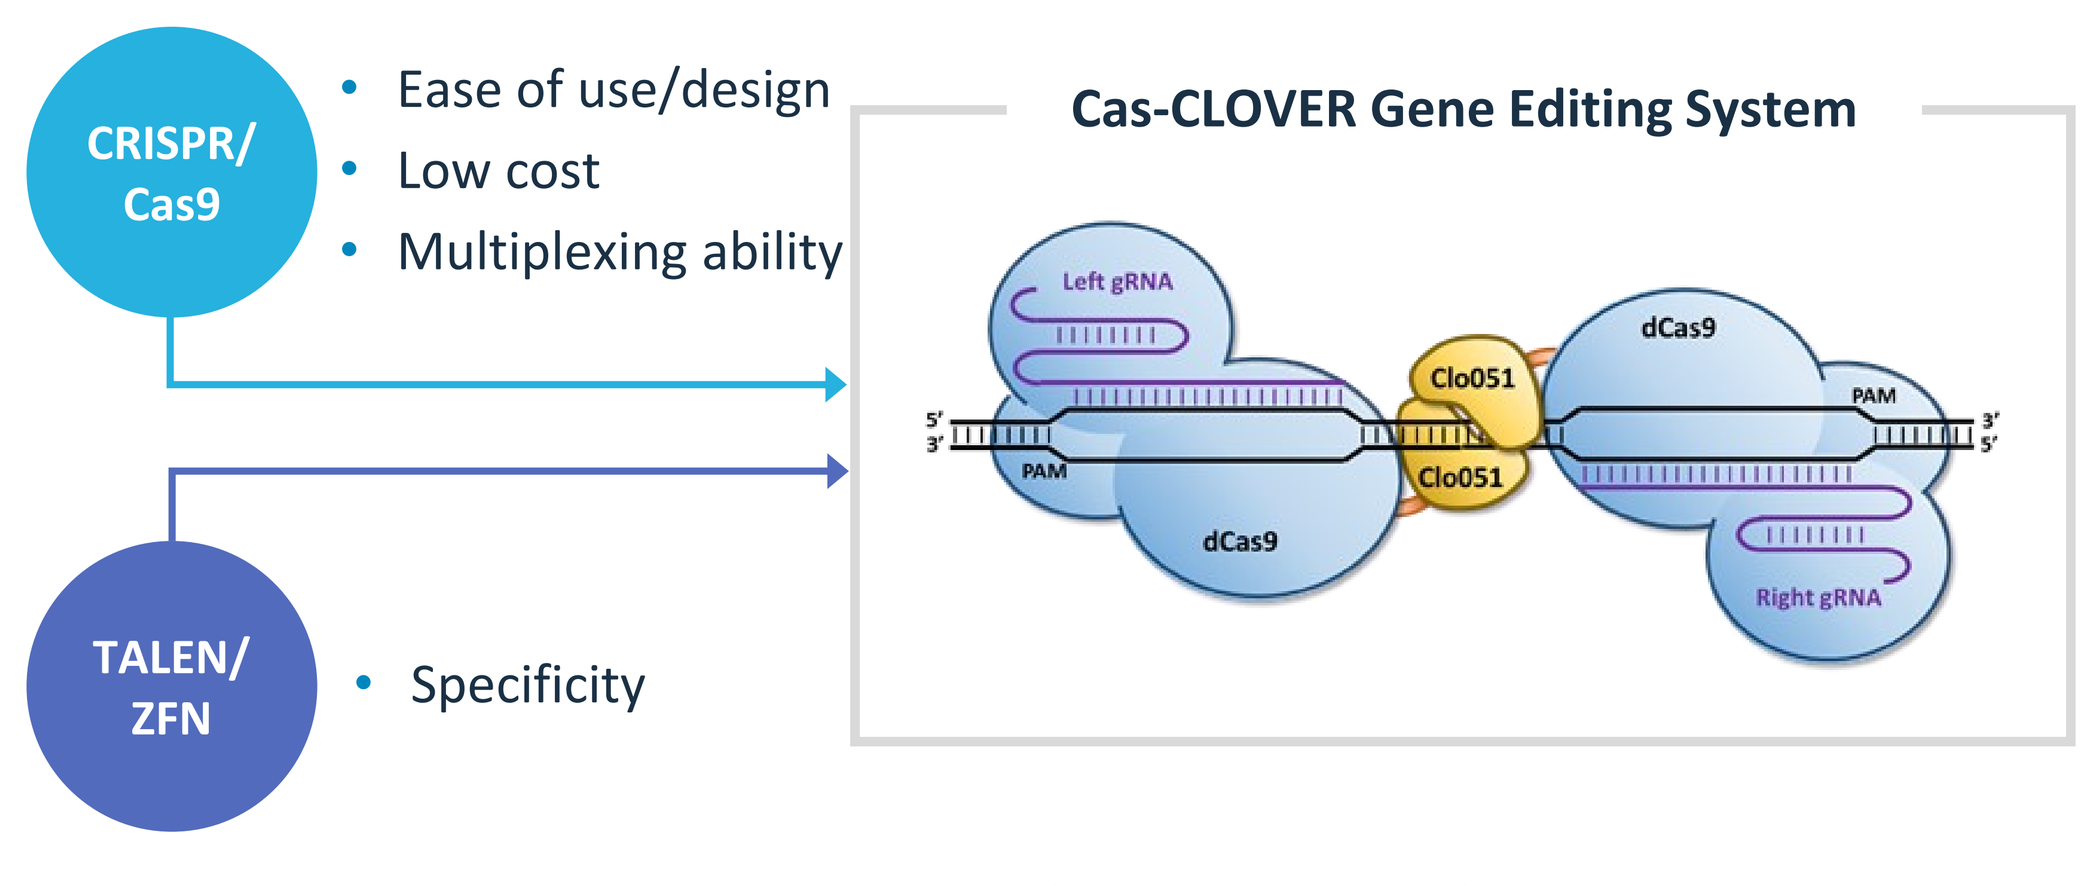
CRISPR/Cas9
Ease of use/design
Low cost
Multiplexing ability
TALEN/ZFN
Specificity
Cas-CLOVER Gene Editing System
Left gRNA
Right gRNA
dCas9
Clo051
PAM

Fig. 3 Cas-CLOVER Gene Editing System combines the strengths of CRISPR/Cas9 and TALEN/ZFN editing, and efficiently edits resting T cells. The site-specific Cas-CLOVER Gene Editing System consists of a dimeric gRNA-guided nuclease. Editing activity is guided by a pair of gRNAs allowing for ease of design as well as multiplexing ability found in CRISPR/Cas9-based systems. At the same time editing activity is contingent on dimerization of two half domains, similar to TALENs or ZFNs, leading to exquisite editing specificity. Cas-CLOVER gene editing is employed to knock out TCR and MHC I in resting T cells.

Depletion of Residual TCR-positive Cells Results in Highly Pure TCR KO P-BCMA-ALLO1

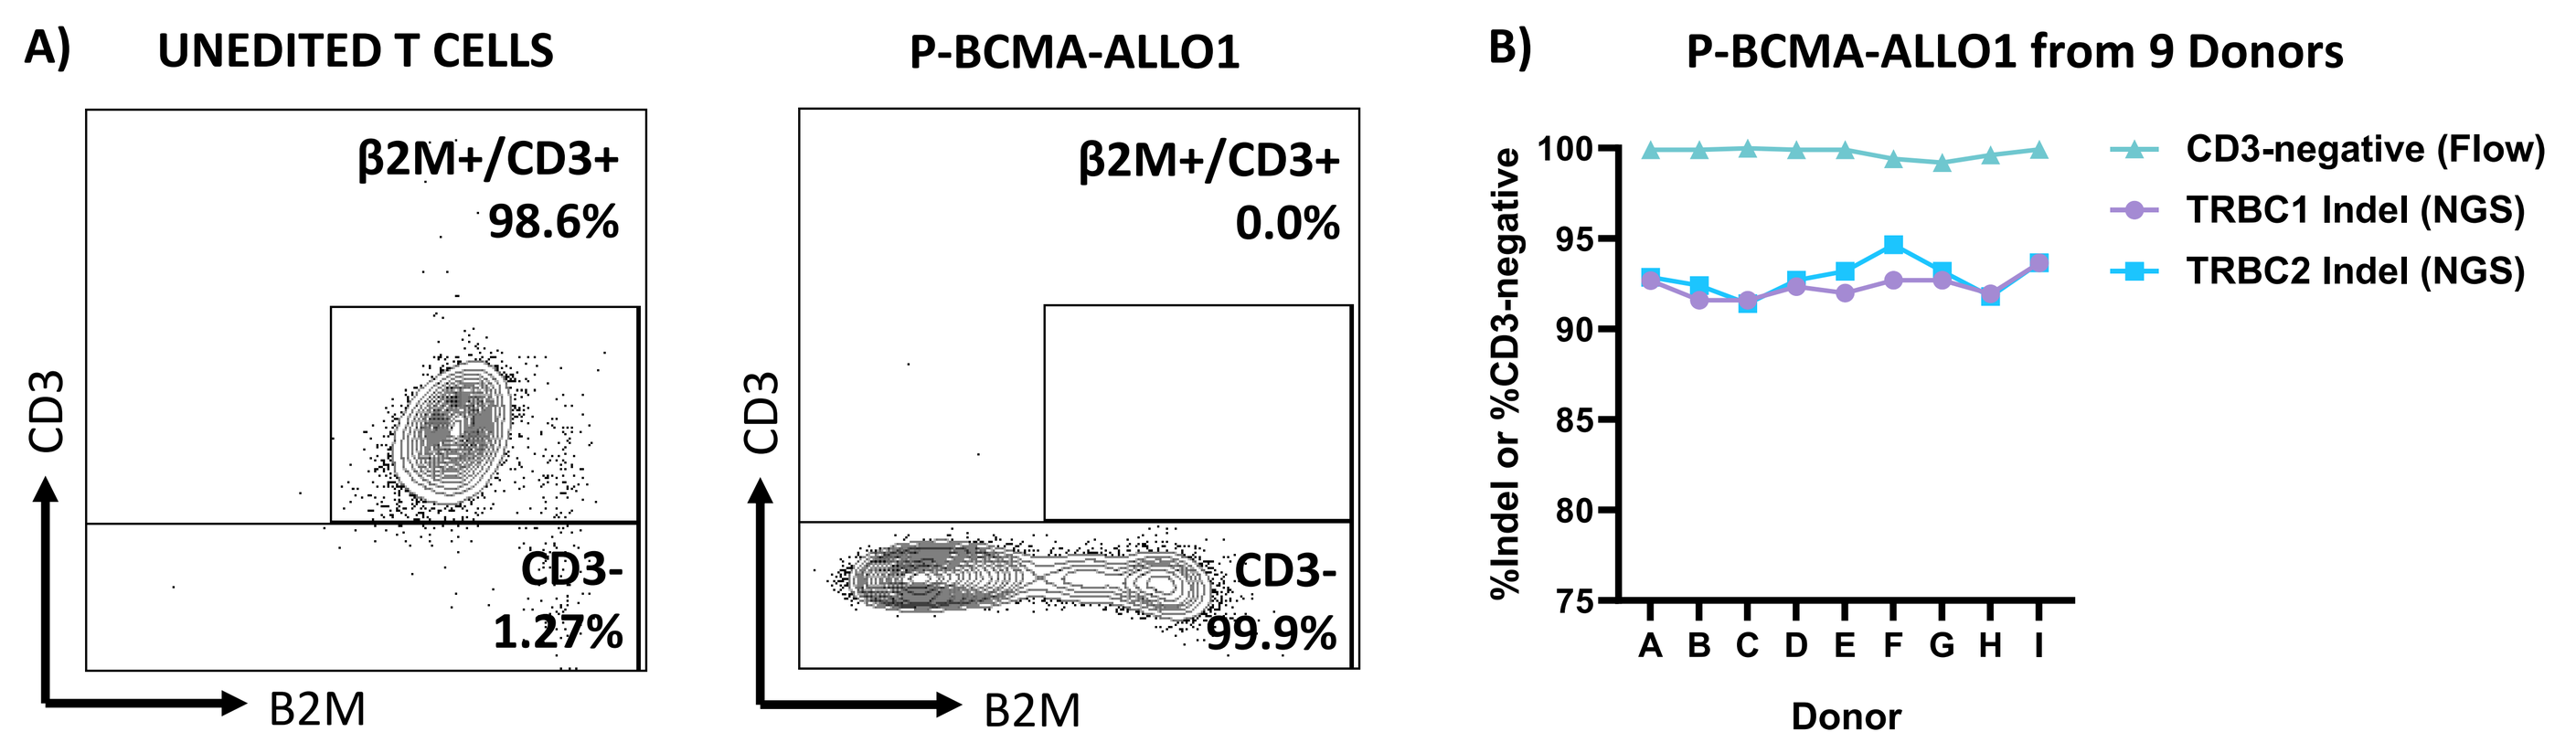
A) UNEDITED T CELLS
P-BCMA-ALLO1
β2M+/CD3+ 98.6%
CD3- 1.27%
β2M+/CD3+ 0.0%
CD3- 99.9%
CD3
B2M
B) P-BCMA-ALLO1 from 9 Donors
CD3-negative (Flow)
TRBC1 Indel (NGS)
TRBC2 Indel (NGS)
%Indel or %CD3-negative
Donor

Fig. 4 Cas-CLOVER gene editing in combination with removal of residual TCR-positive cells leads to high levels of TCR KO in P-BCMA-ALLO1. A) The final P-BCMA-ALLO1 product undergoes robust clinical scale depletion of residual CD3+/TCR+ cells. B) High levels of TCR KO were observed across 9 healthy donors, both by flow cytometry for cell surface CD3 as well as by NGS to quantify indel formation at the TRBC1 and TRBC2 loci. The efficient removal of TCR+ cells generates a safe product that is unable to mediate graft versus host disease (GVHD).

Cas-CLOVER™ Induces Only Low Levels of Off-Target Editing

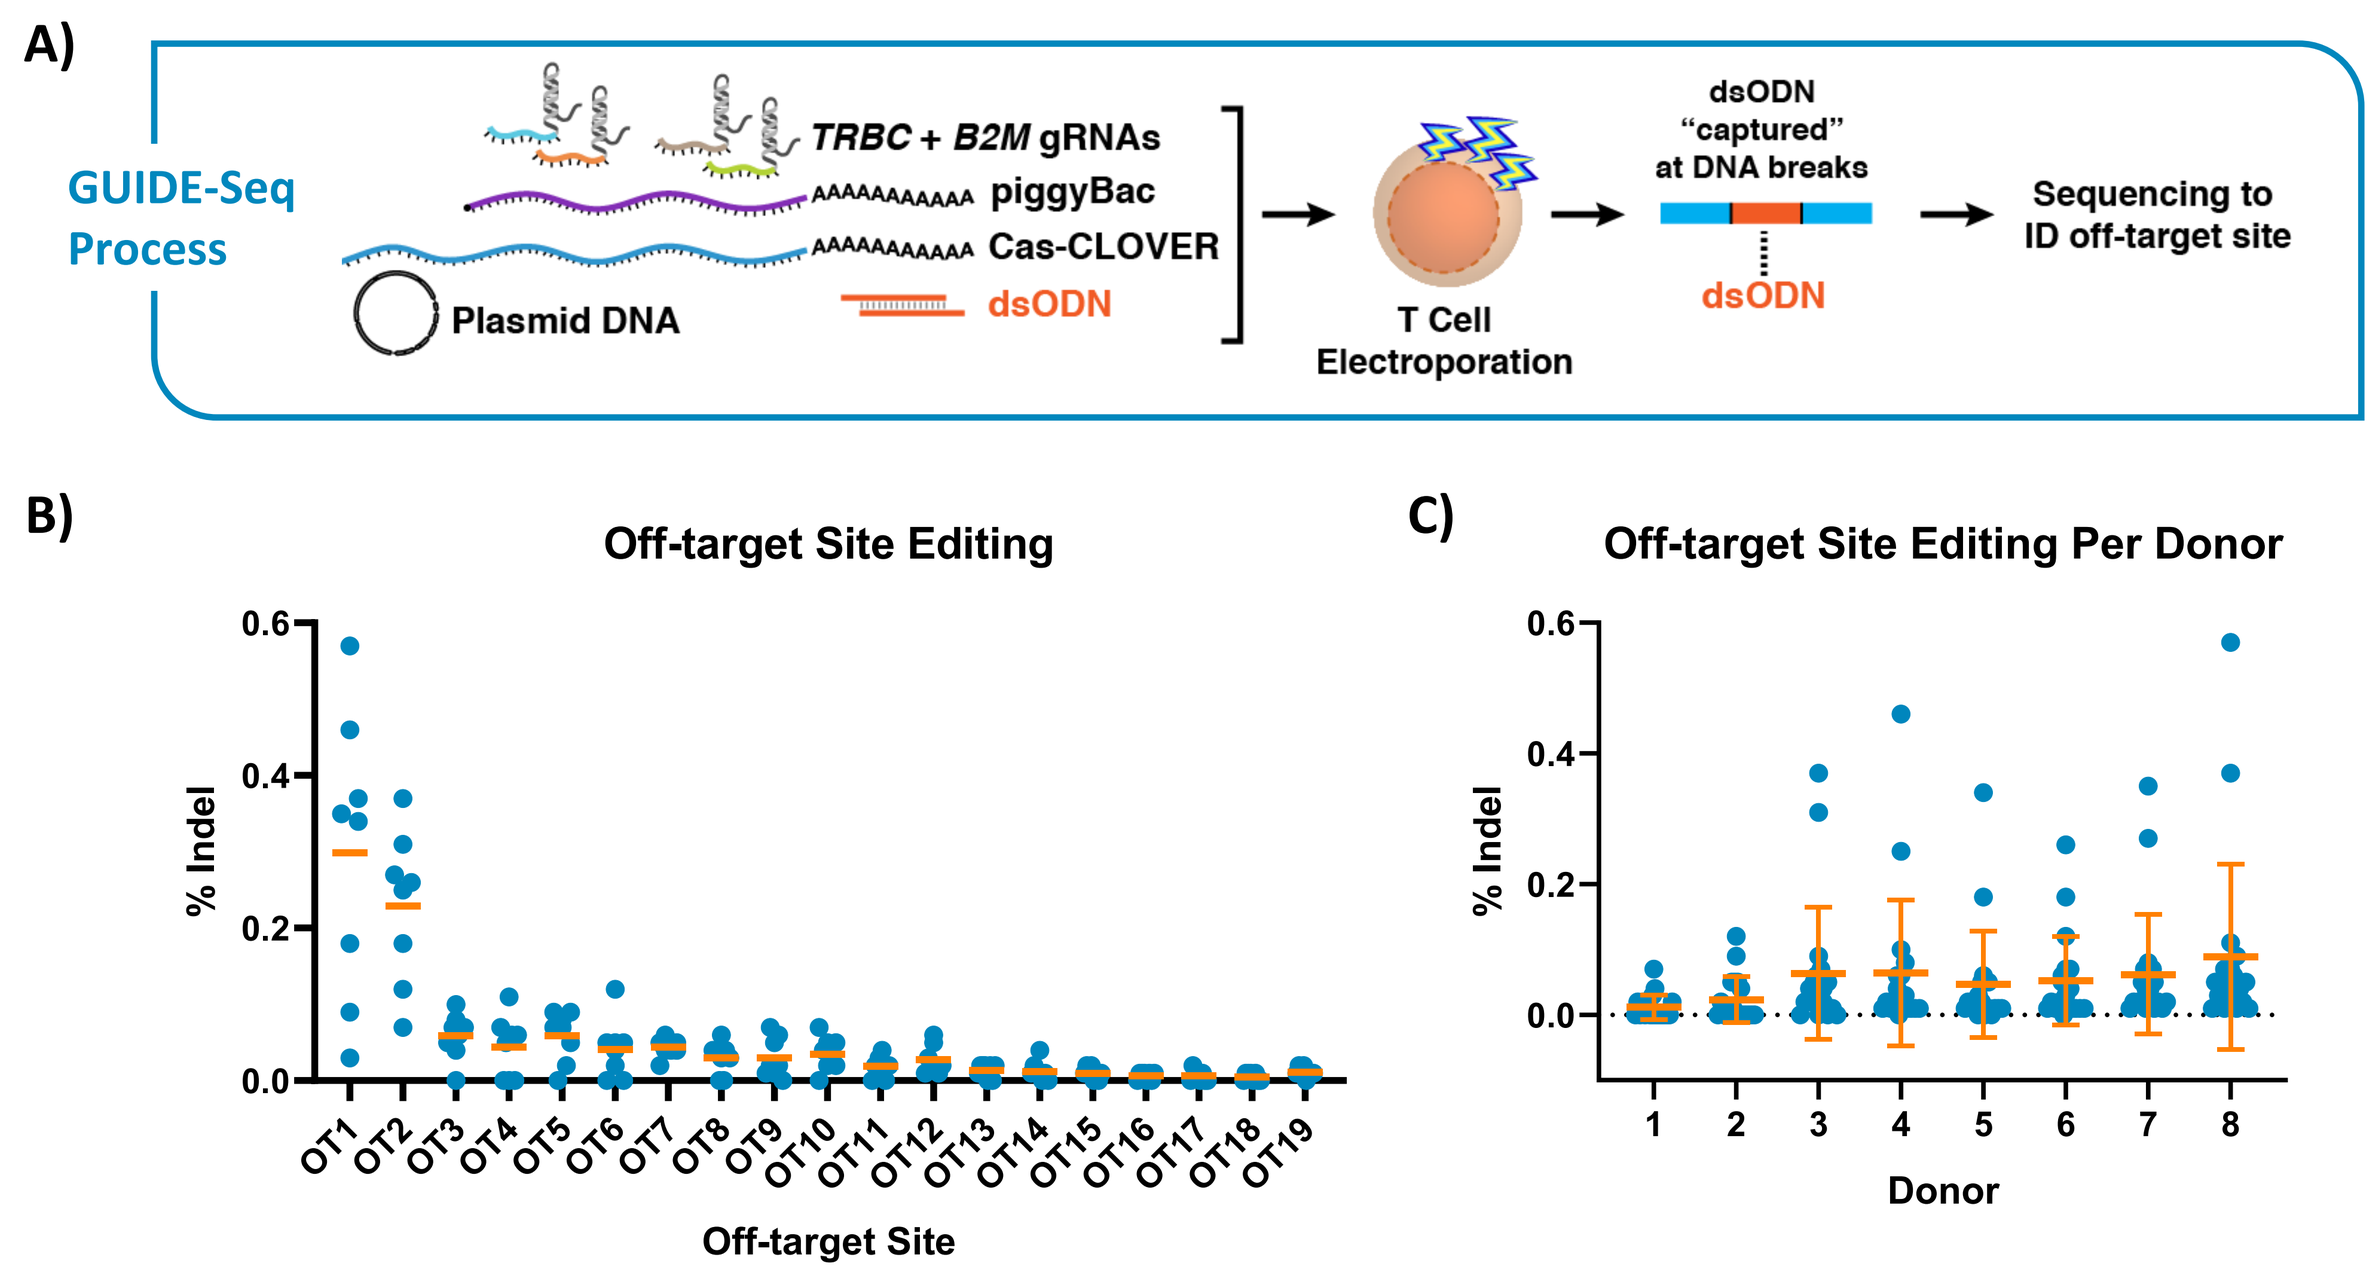
A) GUIDE-Seq Process
TRBC + B2M gRNAs
piggyBac
Cas-CLOVER
Plasmid DNA
dsODN
T Cell Electroporation
dsODN "captured" at DNA breaks
Sequencing to ID off-target site
B) Off-target Site Editing
% Indel
Off-target Site
C) Off-target Site Editing Per Donor
Donor

Fig. 5 Cas-CLOVER exhibits low editing activity at GUIDE-Seq identified off-target sites. A) GUIDE-Seq was used to identify candidate off-target sites for Cas-CLOVER mediated TCR and MHC class I knockout in T cells. Out of a total of 149 candidate off-target sites, 19 bona fide off-target sites were identified via targeted sequencing. B) Indel frequencies observed at the 19 identified off-target sites in P-BCMA-ALLO1 produced from 8 healthy donors. Dots represent individual donors; orange bars indicate the mean. C) Average off-target editing rates across all off-target editing sites observed in P-BCMA-ALLO1 produced from 8 healthy donors. Dots represent individual off-target editing sites; orange bars indication mean ± SD indel frequencies.

Booster Molecule Allows for Hundreds of Doses of T_{SCM}-rich P-BCMA-ALLO1 from a Single Manufacturing Run

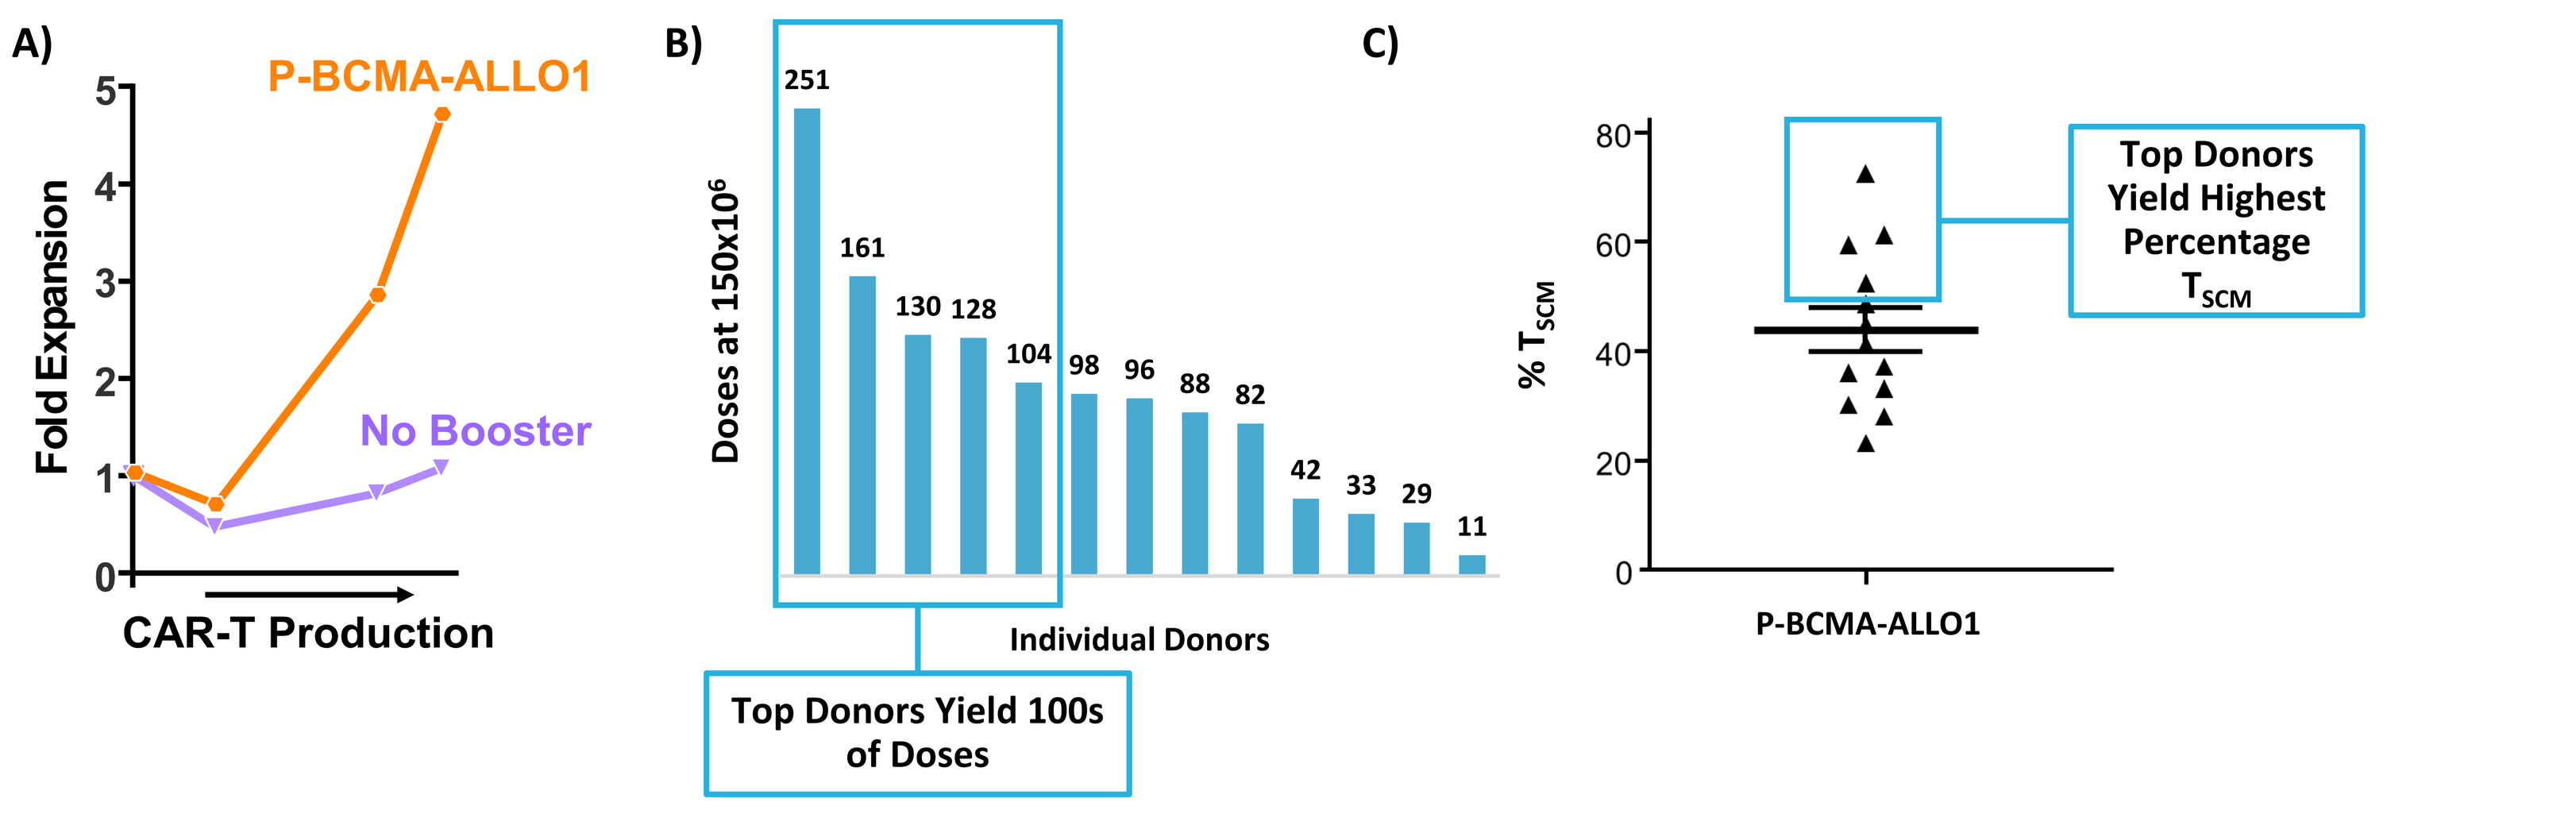
A)
P-BCMA-ALLO1
No Booster
Fold Expansion
CAR-T Production
B)
Doses at 150x10⁶
251
161
130
128
104
98
96
88
82
42
33
29
11
Individual Donors
Top Donors Yield 100s of Doses
C)
% T_{SCM}
Top Donors Yield Highest Percentage T_{SCM}
P-BCMA-ALLO1

Fig. 6 The Booster Molecule increases the yield of P-BCMA-ALLO1 cells while maintaining the desirable early memory phenotype. A) P-BCMA-ALLO1 CAR-T cells were produced in presence or absence of the Booster Molecule. Addition of the Booster Molecule led to significant increases in cell expansion during production. B) Use of the Booster Molecule for P-BCMA-ALLO1 production allows the generation of a high number of patient doses from a single manufacturing run. C) At the same time, P-BCMA-ALLO1 produced using the Booster Molecule preserves a product phenotype rich in T_{SCM} cells.

P-BCMA-ALLO1 Shows Robust Effector Function and High Proliferative Capacity In Vitro

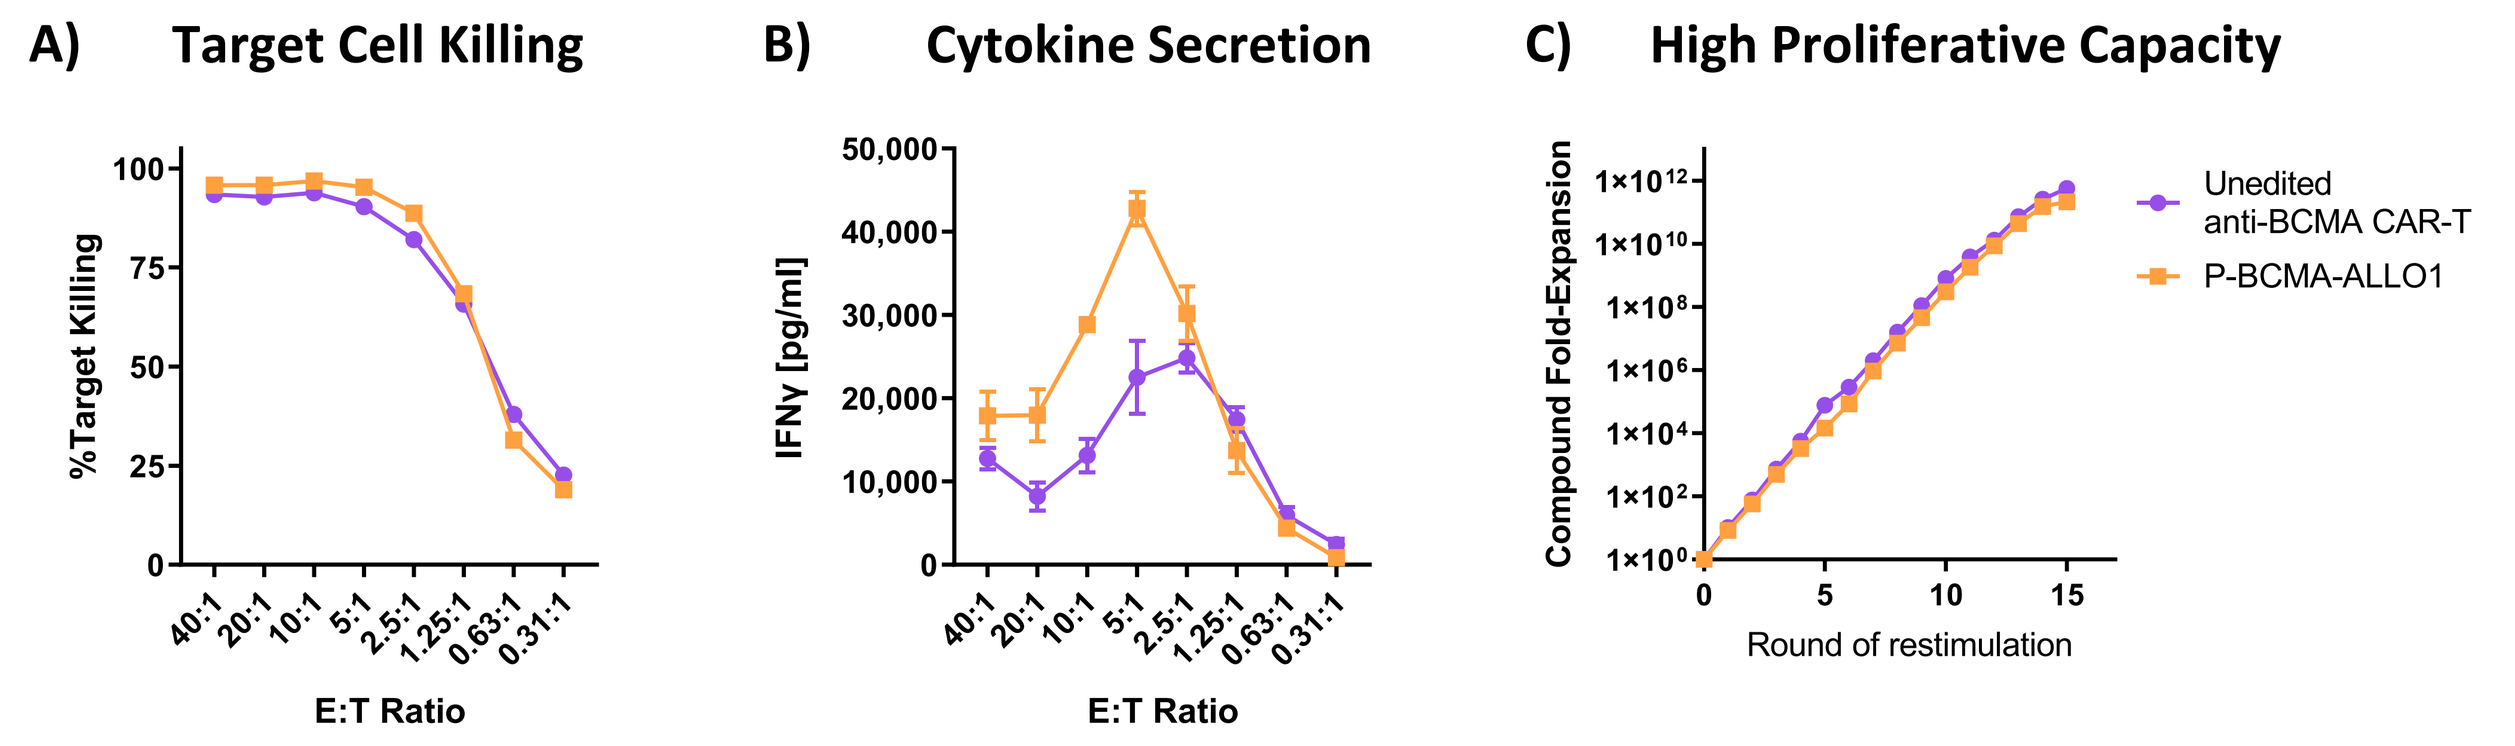
A) Target Cell Killing
%Target Killing
E:T Ratio
B) Cytokine Secretion
IFNγ [pg/ml]
E:T Ratio
C) High Proliferative Capacity
Compound Fold-Expansion
Round of restimulation
Unedited anti-BCMA CAR-T
P-BCMA-ALLO1

Fig. 7 P-BCMA-ALLO1 exhibits potent, target-specific effector function in vitro. P-BCMA-ALLO1 exhibited efficient target-dependent cell killing (A) and IFNγ cytokine secretion (B). In an in vitro serial restimulation assay, P-BCMA-ALLO1 exhibited a high level of proliferative capacity. Both the in vitro effector function and proliferative capacity of P-BCMA-ALLO1 were comparable to that observed in unedited TCR+ anti-BCMA CAR-T cells.

P-BCMA-ALLO1 Exhibits Potent Anti-Tumor Activity In Vivo

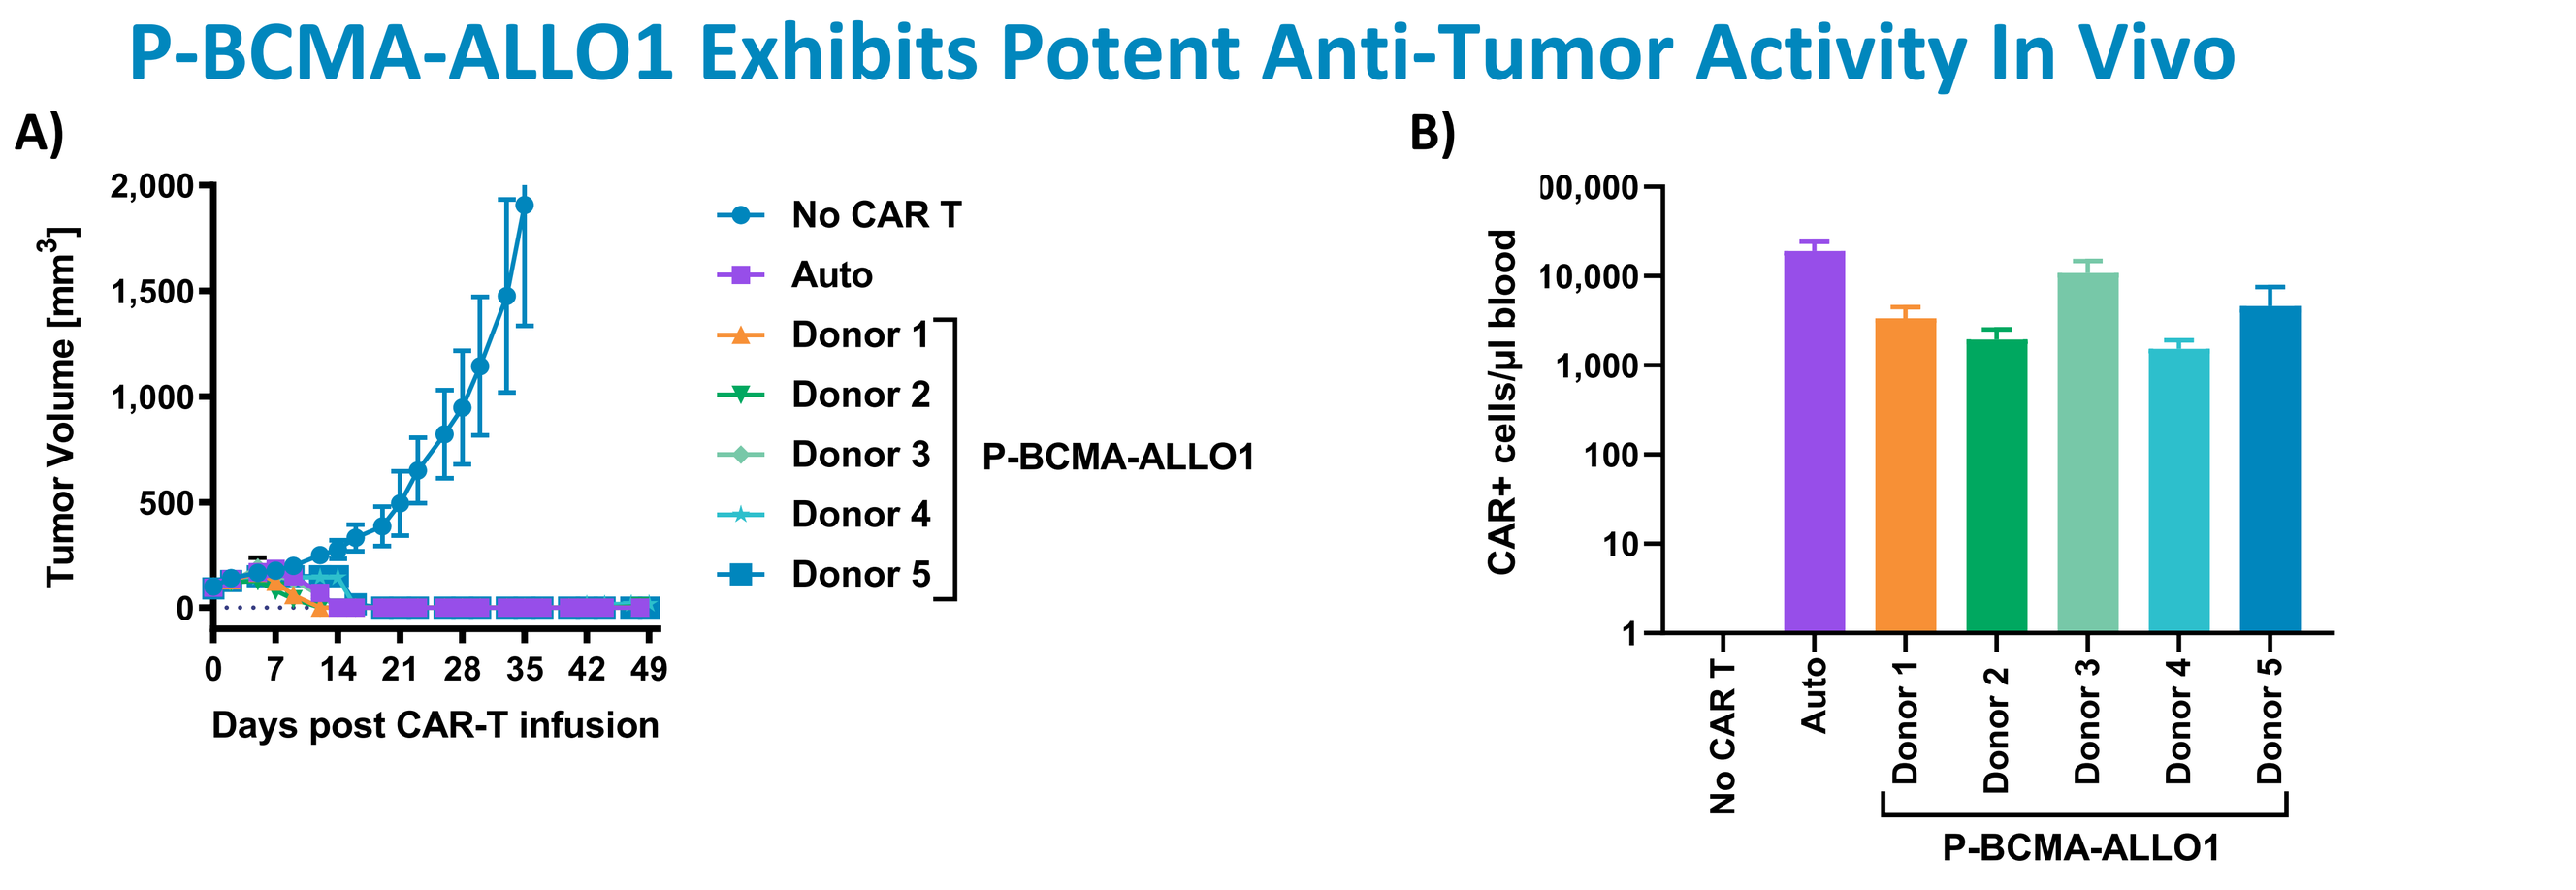
A)
Tumor Volume [mm³]
Days post CAR-T infusion
No CAR T
Auto
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
P-BCMA-ALLO1
B)
CAR+ cells/µl blood
No CAR T
Auto
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
P-BCMA-ALLO1

Fig. 8 P-BCMA-ALLO1 exhibits potent anti-tumor activity in the RPMI-8226 in vivo model of MM. P-BCMA-ALLO1 produced from 5 donors was tested alongside unedited anti-BCMA CAR-T cells ("Auto", CAR-positive & TCR-positive) in RPMI-8226 tumor bearing NSG mice. A) Tumor volume of subcutaneous masses was measured via caliper. P-BCMA-ALLO1 exhibited potent and sustained anti-tumor efficacy leading to complete tumor regression in all 5 donors. B) P-BCMA-ALLO1 demonstrated robust in vivo expansion in response to tumor. Circulating P-BCMA-ALLO1 cells were quantified by flowcytometry in the blood of treated animals. Peak levels of circulating CAR-T cells at Day 14 after CAR-T infusion are shown.

P-BCMA-ALLO1 Exhibits Tumor Control and Durability in an Aggressive Systemic MM.1S In Vivo Model

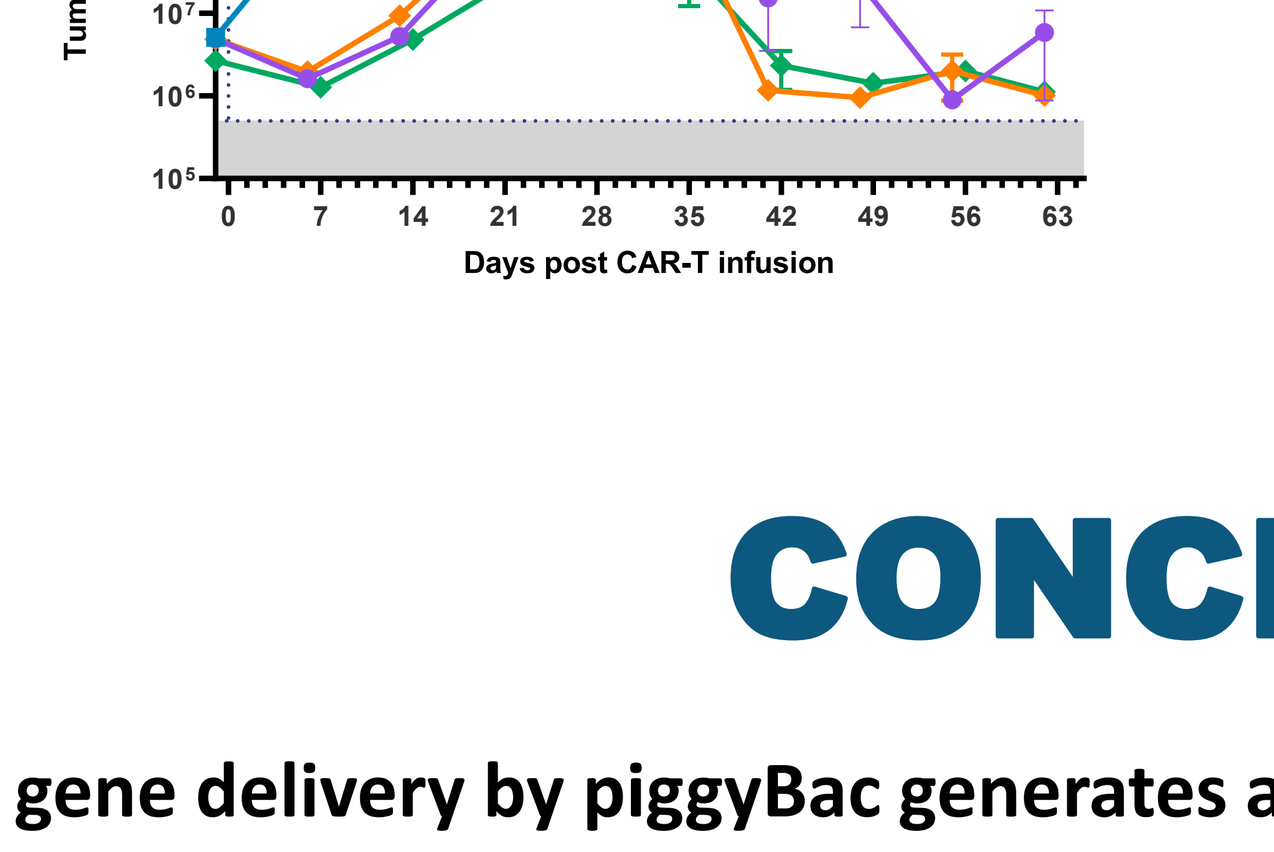
Tumor Bioluminescence (photons/sec)
Days post CAR-T infusion
No CAR-T
Unedited anti-BCMA CAR-T
P-BCMA-ALLO1 - Donor A
P-BCMA-ALLO1 - Donor B

Fig. 9 P-BCMA-ALLO1 induced durable tumor control in the systemic MM.1S mouse model of MM and exhibited the ability to spontaneously recontrol the tumor. P-BCMA-ALLO1 from 2 healthy donors was used to treat a systemic MM.1S tumor model in NSG mice. Tumor burden was measured via bioluminescence imaging. Following P-BCMA-ALLO1 infusion, tumor control was observed. Further, upon spontaneous relapse of the tumor, P-BCMA-ALLO1 was able to recontrol the tumor and prevent further relapse. This demonstrates the durability of the T_{SCM} rich P-BCMA-ALLO1 allogeneic CAR-T.

CONCLUSIONS

- **Nonviral gene delivery by piggyBac generates a CAR-T product rich in desirable T_{SCM} cells**
- **Cas-CLOVER efficiently gene edits resting T cells with high fidelity and low levels of off-target editing**
- **Booster molecule allows to generate hundreds of doses of allogeneic CAR-T per manufacturing run**
- **P-BCMA-ALLO1 exhibits potent effector function in vitro and effective and durable anti-tumor activity in vivo**
- **Results support rapid advancement of P-BCMA-ALLO1 into the clinic for treatment of MM**